ABSTRACT/POSTER NUMBER B06

CB-020, an Induced Pluripotent Stem Cell (iPSC)-Derived Allogeneic CAR-NK Cell Therapy, Engineered for Enhanced Activity Against Solid Tumors

Rodolfo Gonzalez[1], Erica Hennessy[1], Zachary Pappalardo[1], Jackie Bouza[1], Varsha Ravi[1], Christian Sunderland[1], Chae Lee[1], Sushma Ram[1], Devin Mutha[1], Raymond Guo[1], Art Aviles[1], Leslie Edwards[1], Paul Donohoue[1], Tristan Fowler[1], Zili An[1], and Steven Kanner[1]
[1]Caribou Biosciences, Inc., Berkeley, California, USA

Background

Chimeric antigen receptor (CAR)-T cell therapies generally have not demonstrated robust and reproducible antitumor activity against solid malignancies. Natural killer (NK) cells express the receptor NKG2D promoting their innate ability to detect and kill tumors that express stress ligands (MICA, MICB, and ULBPs). CB-020, an iPSC-derived CAR-NK cell therapy, is being developed for targeting solid tumors and their associated metastases. ROR1 is a receptor tyrosine-kinase-like antigen expressed in a wide range of tumors including ovarian, lung, pancreatic, and triple negative breast cancer, and it drives tumor cell growth, survival, and metastasis. CB-020 is being engineered to express a ROR1-specific CAR (ROR1-CAR) to enhance its specificity and function and promote trafficking to ROR1⁺ tumors. Additionally, CAR expression will address the heterogeneity of antigen expression on solid tumors, enabling CB-020 to detect tumor cells via its native NKG2D receptors and the ROR1 CAR.

iPSC-derived NK cell (iNK) platform

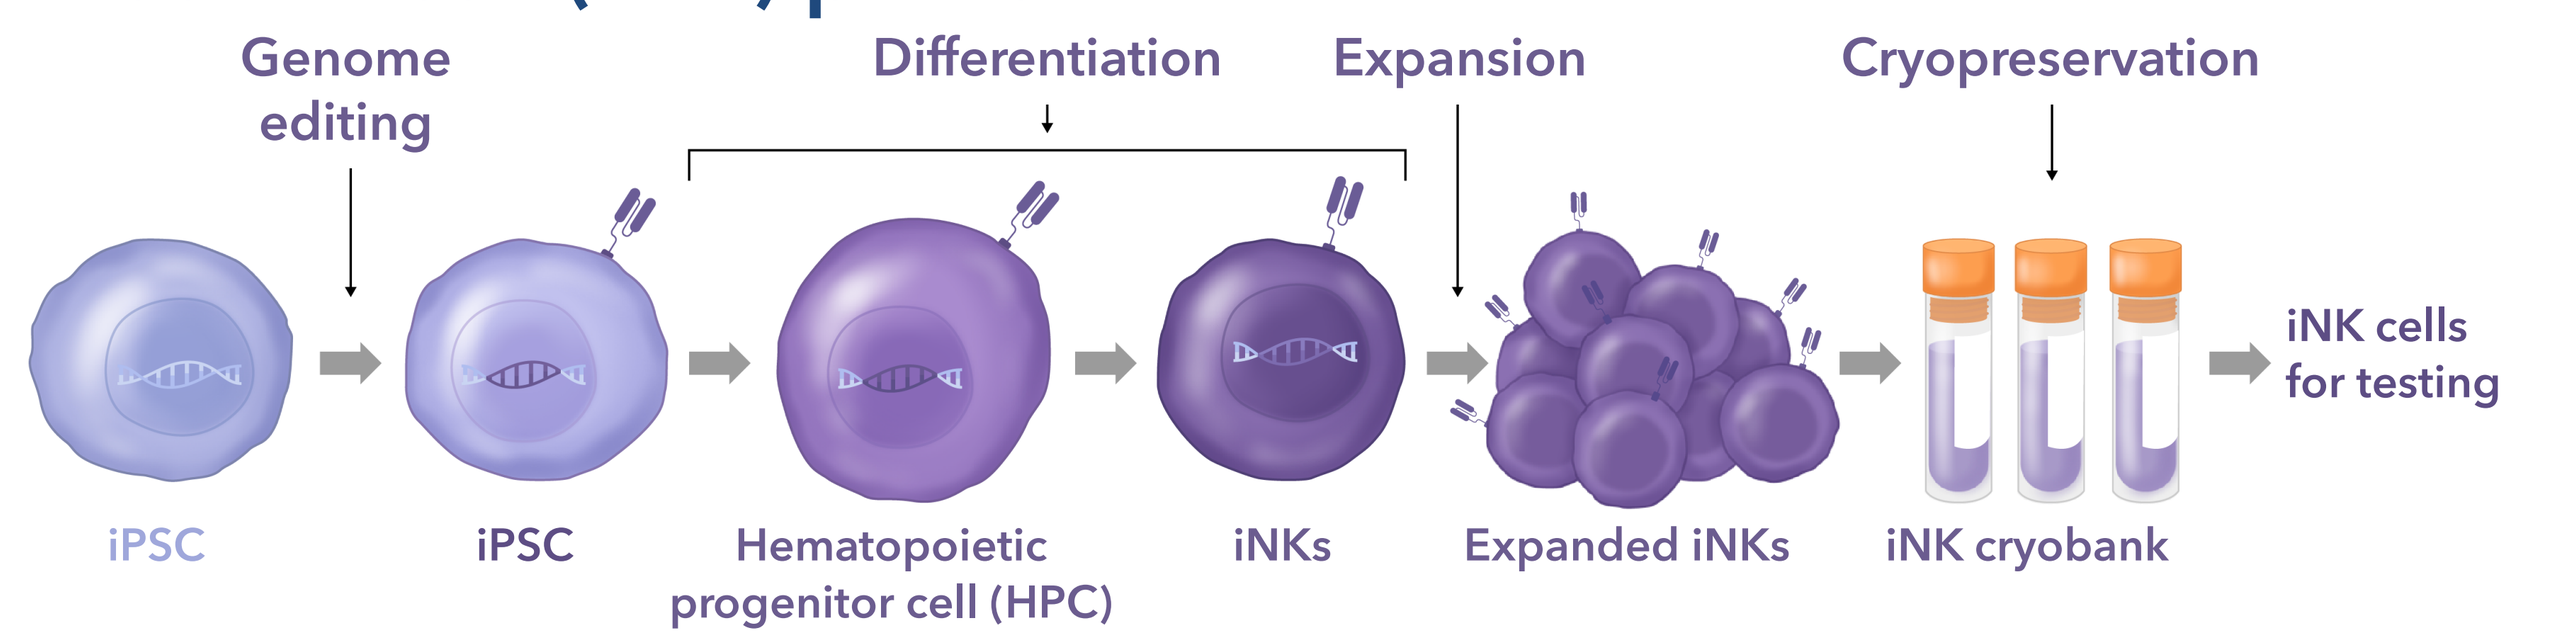
Genome editing
Differentiation
Expansion
Cryopreservation
iPSC
iPSC
Hematopoietic progenitor cell (HPC)
iNKs
Expanded iNKs
iNK cryobank
iNK cells for testing

Fig 1. Schematic of iPSC editing and differentiation process to edited iNKs. Genome editing is performed at the iPSC stage. Engineered iPSCs are differentiated through hematopoietic progenitor cells to iNKs. Differentiated iNKs are expanded and cryopreserved for subsequent *in vitro* and *in vivo* analysis.

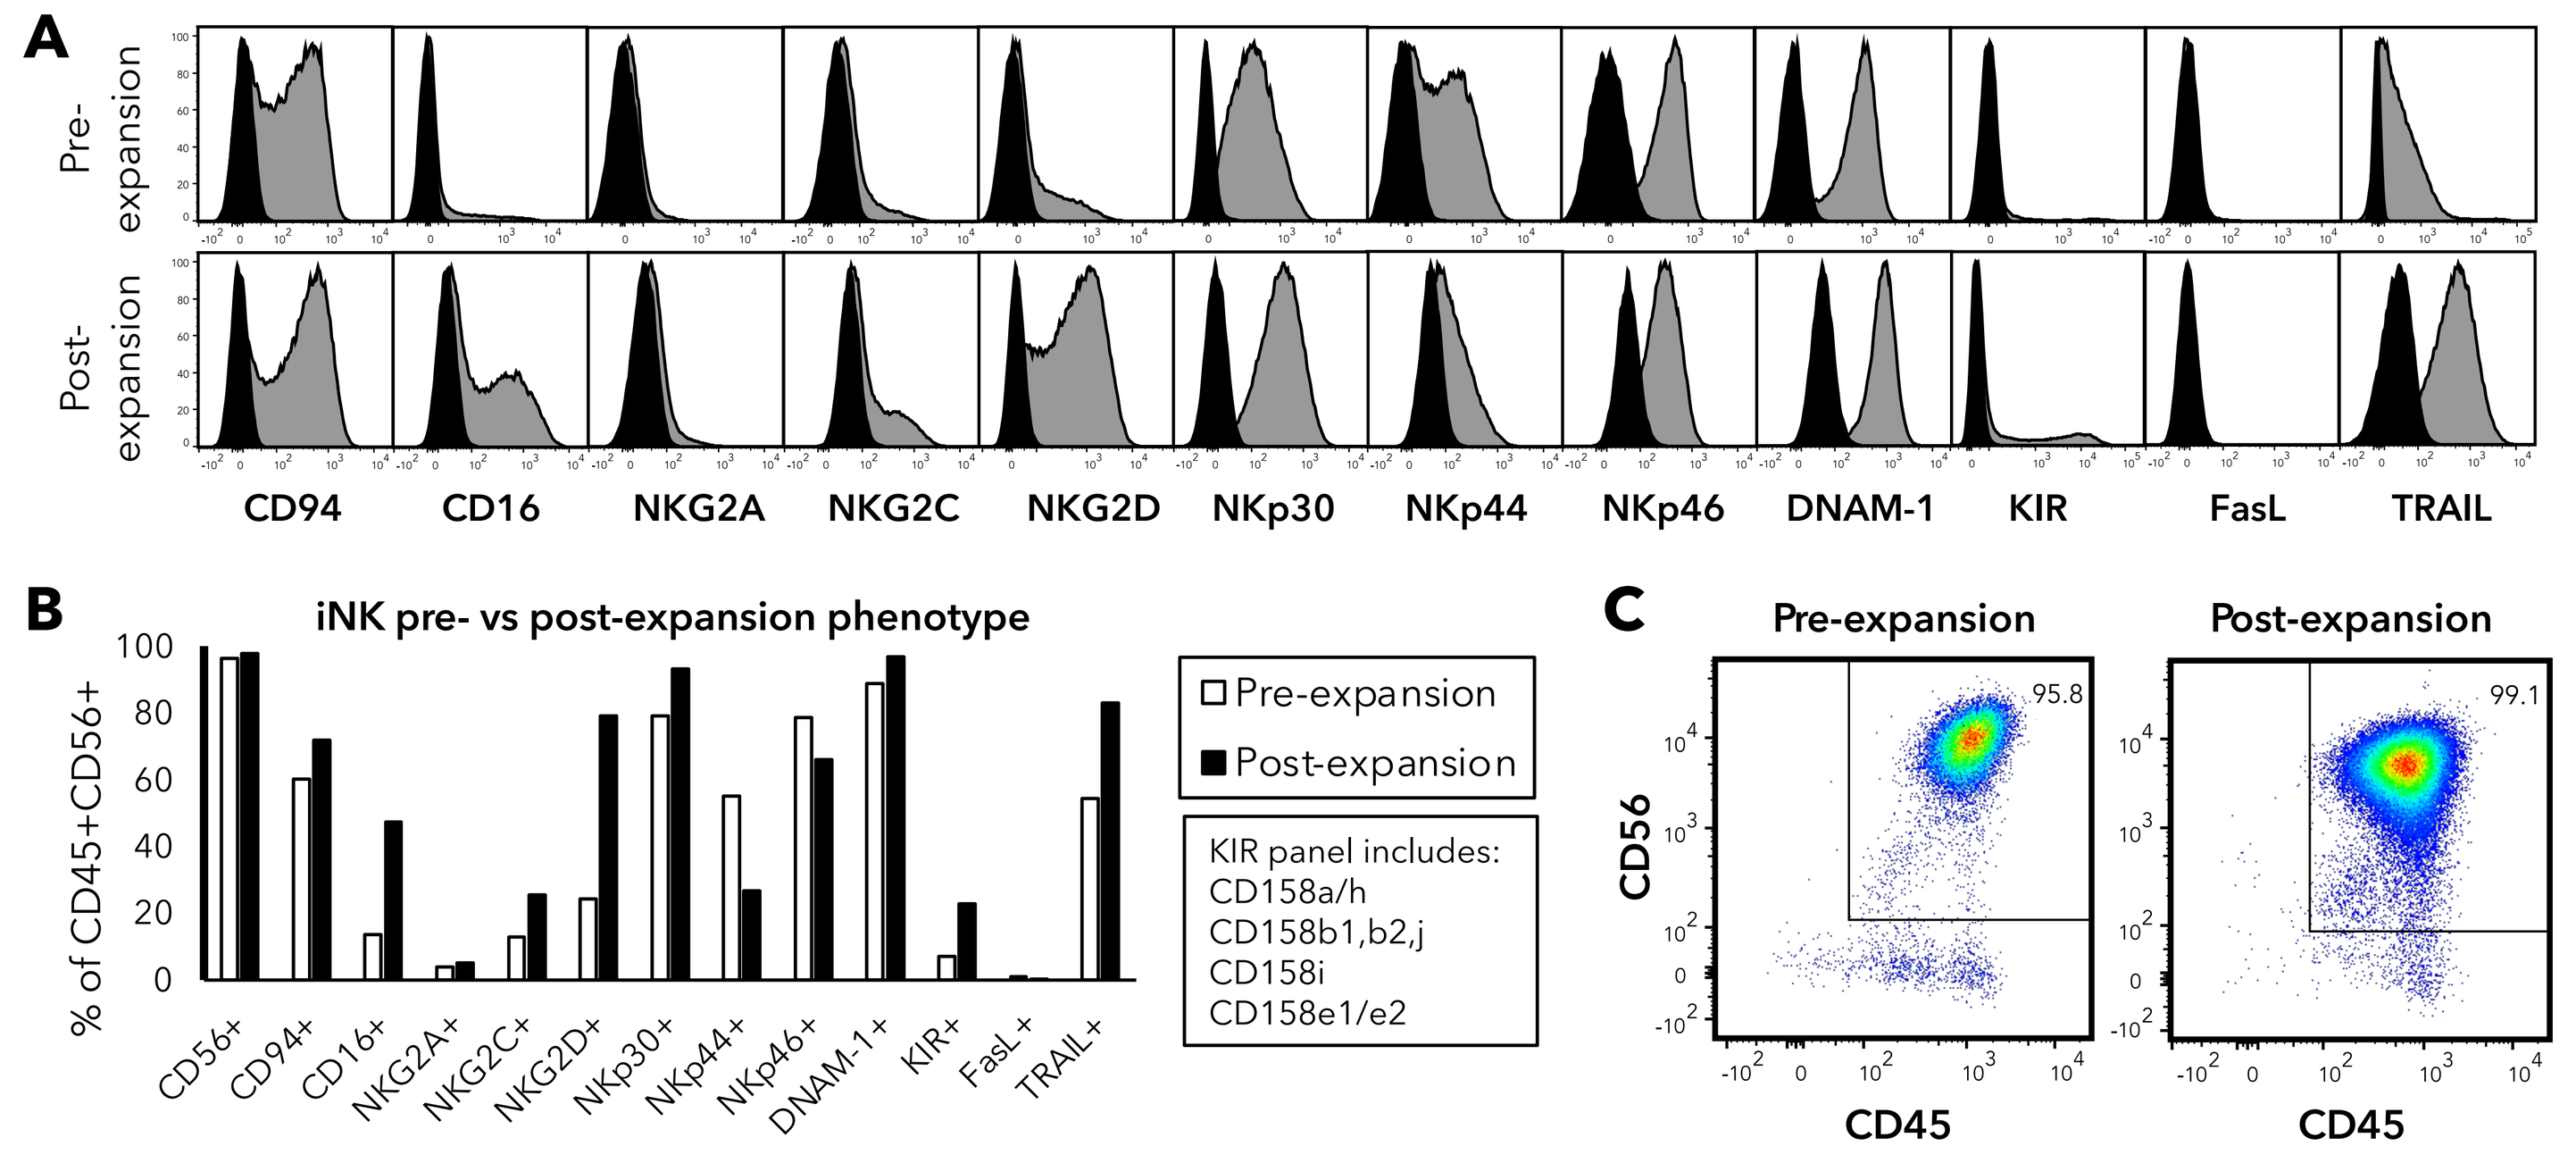
A
Pre-expansion
Post-expansion
CD94 CD16 NKG2A NKG2C NKG2D NKp30 NKp44 NKp46 DNAM-1 KIR FasL TRAIL
B
iNK pre- vs post-expansion phenotype
% of CD45+CD56+
Pre-expansion
Post-expansion
KIR panel includes:
CD158a/h
CD158b1,b2,j
CD158i
CD158e1/e2
C
Pre-expansion
Post-expansion
CD56
CD45
95.8
99.1

Fig 2. iNK pre- and post-expansion phenotype. Increased expression of NK activating receptors post-expansion stage compared to pre-expansion.
(**A**) Expression profile of NK activating receptors (CD16, NKG2C, NKG2D, NKp30, NKp44, NKp46, DNAM-1), inhibitory receptor (NKG2A), effector molecules (FasL and TRAIL) and KIR (pool of activating and inhibitory KIRs).
(**B**) NK marker expression as a percent of CD56+CD45+ cells.
(**C**) iNK purity pre- and post-expansion measure by CD56 and CD45 staining.

ROR1 tumor target and CAR construct

Fig 3. Functional domains of ROR1

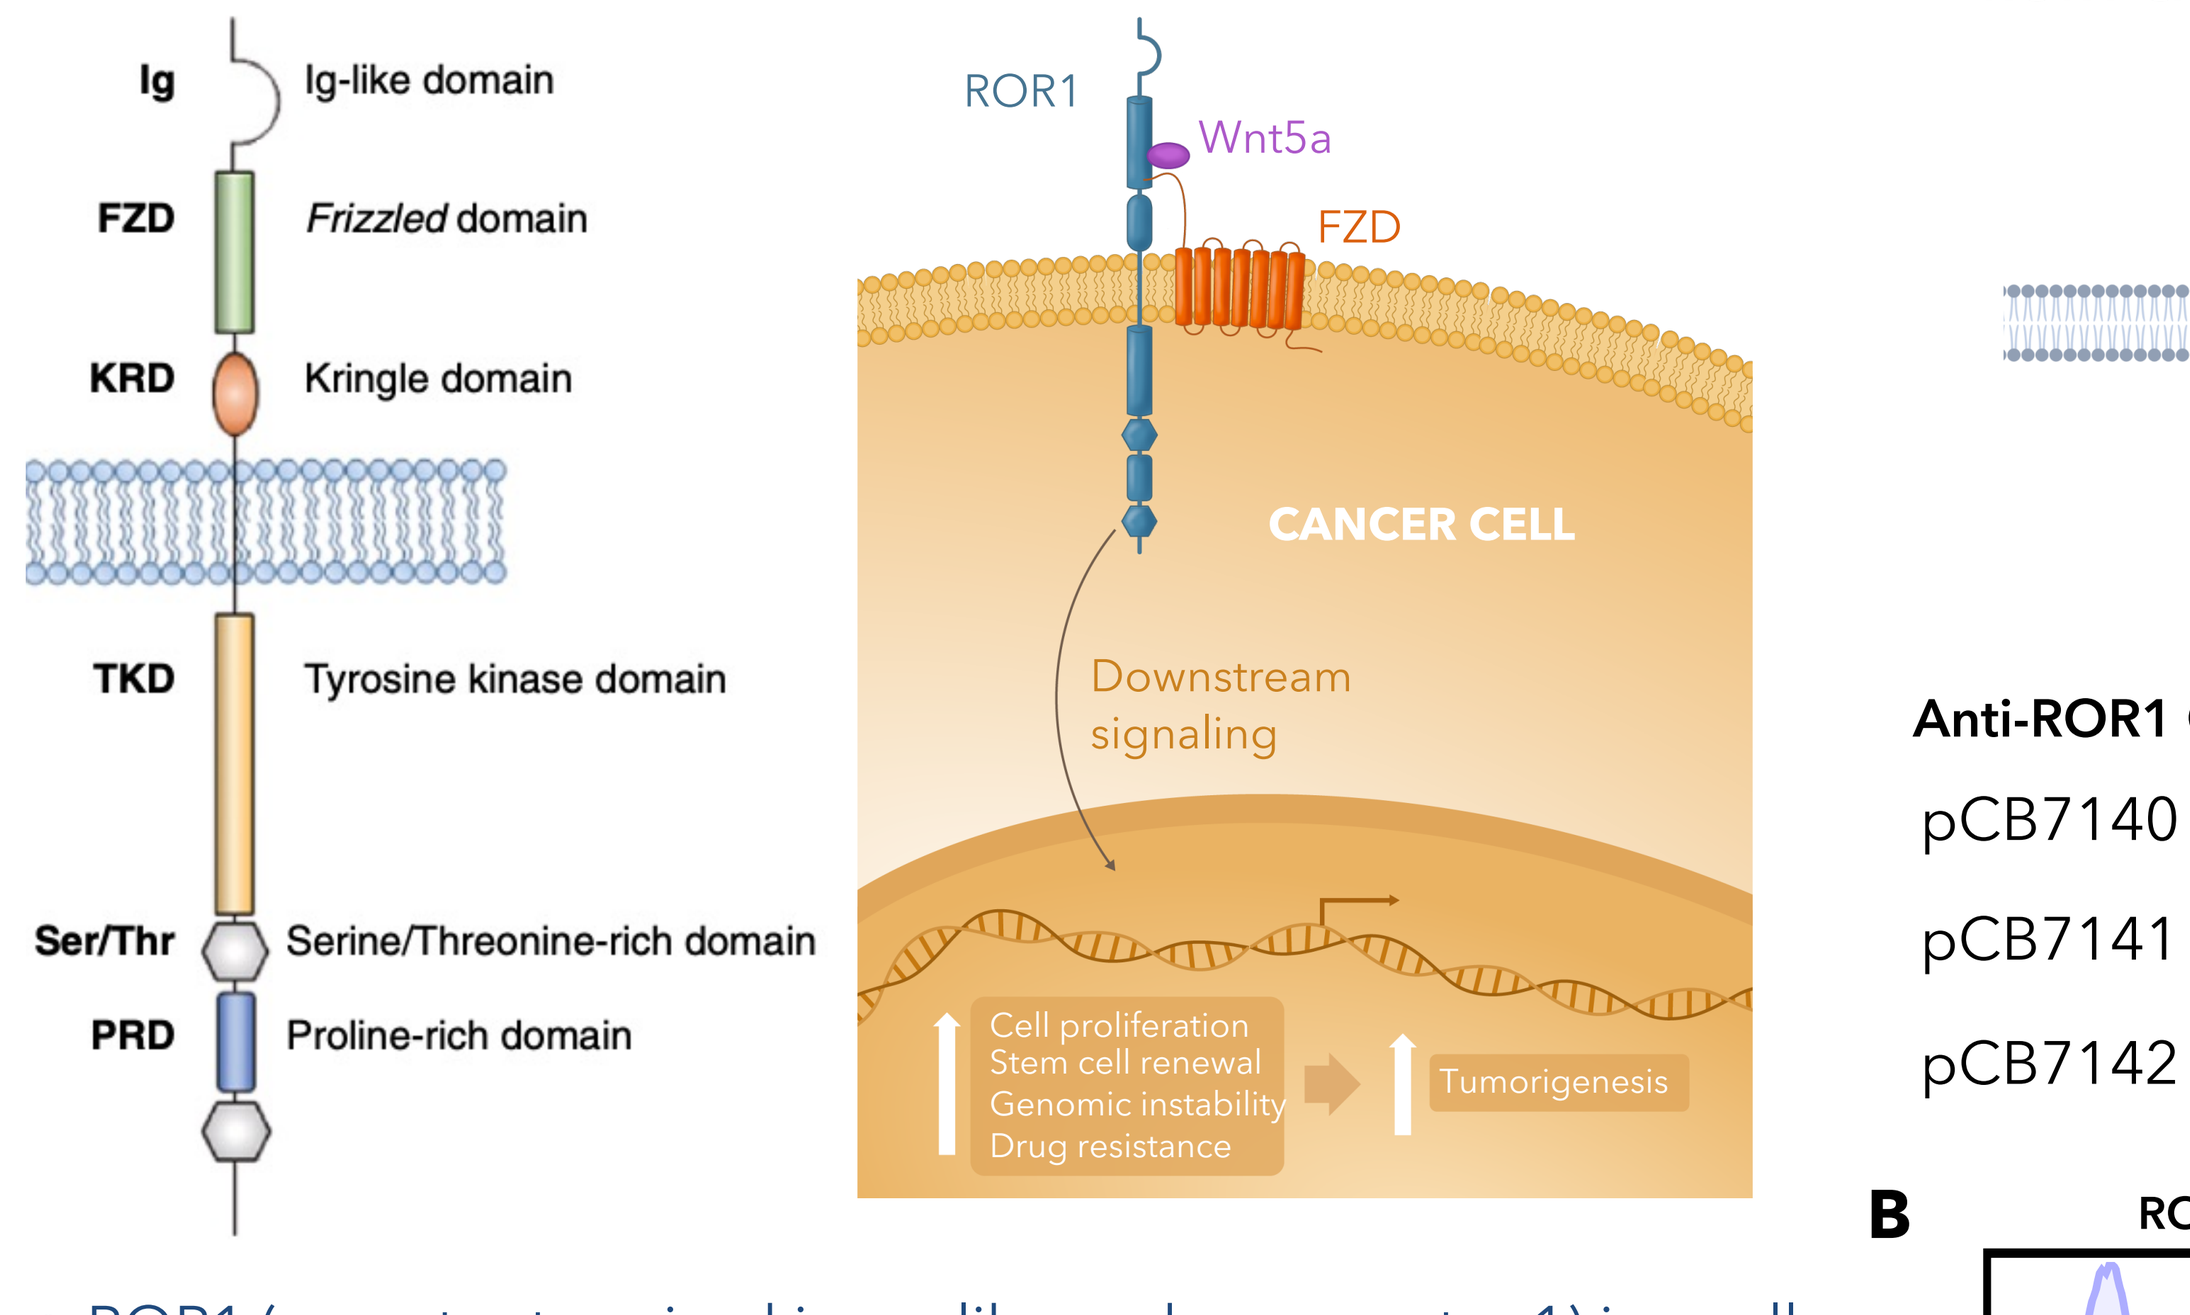
Ig Ig-like domain
FZD Frizzled domain
KRD Kringle domain
TKD Tyrosine kinase domain
Ser/Thr Serine/Threonine-rich domain
PRD Proline-rich domain
ROR1
Wnt5a
FZD
CANCER CELL
Downstream signaling
Cell proliferation
Stem cell renewal
Genomic instability
Drug resistance
Tumorigenesis

- ROR1 (receptor tyrosine kinase-like orphan receptor 1) is a cell surface receptor for Wnt5a
- Wnt5a-ROR1 signaling leads to activation of Rac/Rho pathways for cell motility and invasion
- Cancer expression: ovarian, triple negative breast, lung, colorectal, pancreatic, B cell malignancies
- Normal tissue expression: parathyroid, adipocytes, GI tract, islets, B cell precursors

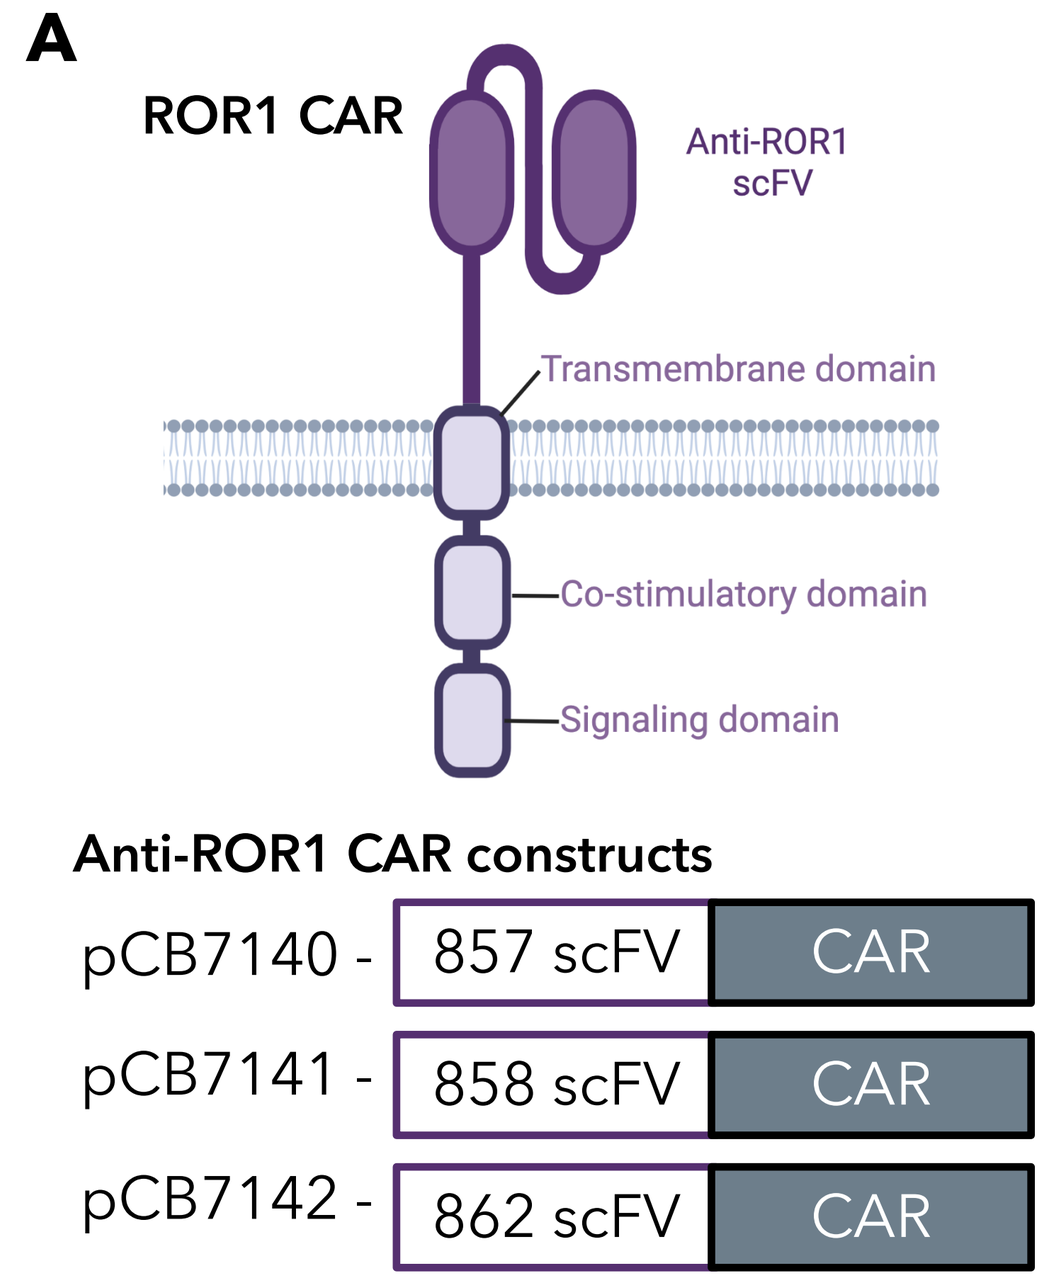
A
ROR1 CAR
Anti-ROR1 scFV
Transmembrane domain
Co-stimulatory domain
Signaling domain
Anti-ROR1 CAR constructs
pCB7140 - 857 scFV CAR
pCB7141 - 858 scFV CAR
pCB7142 - 862 scFV CAR

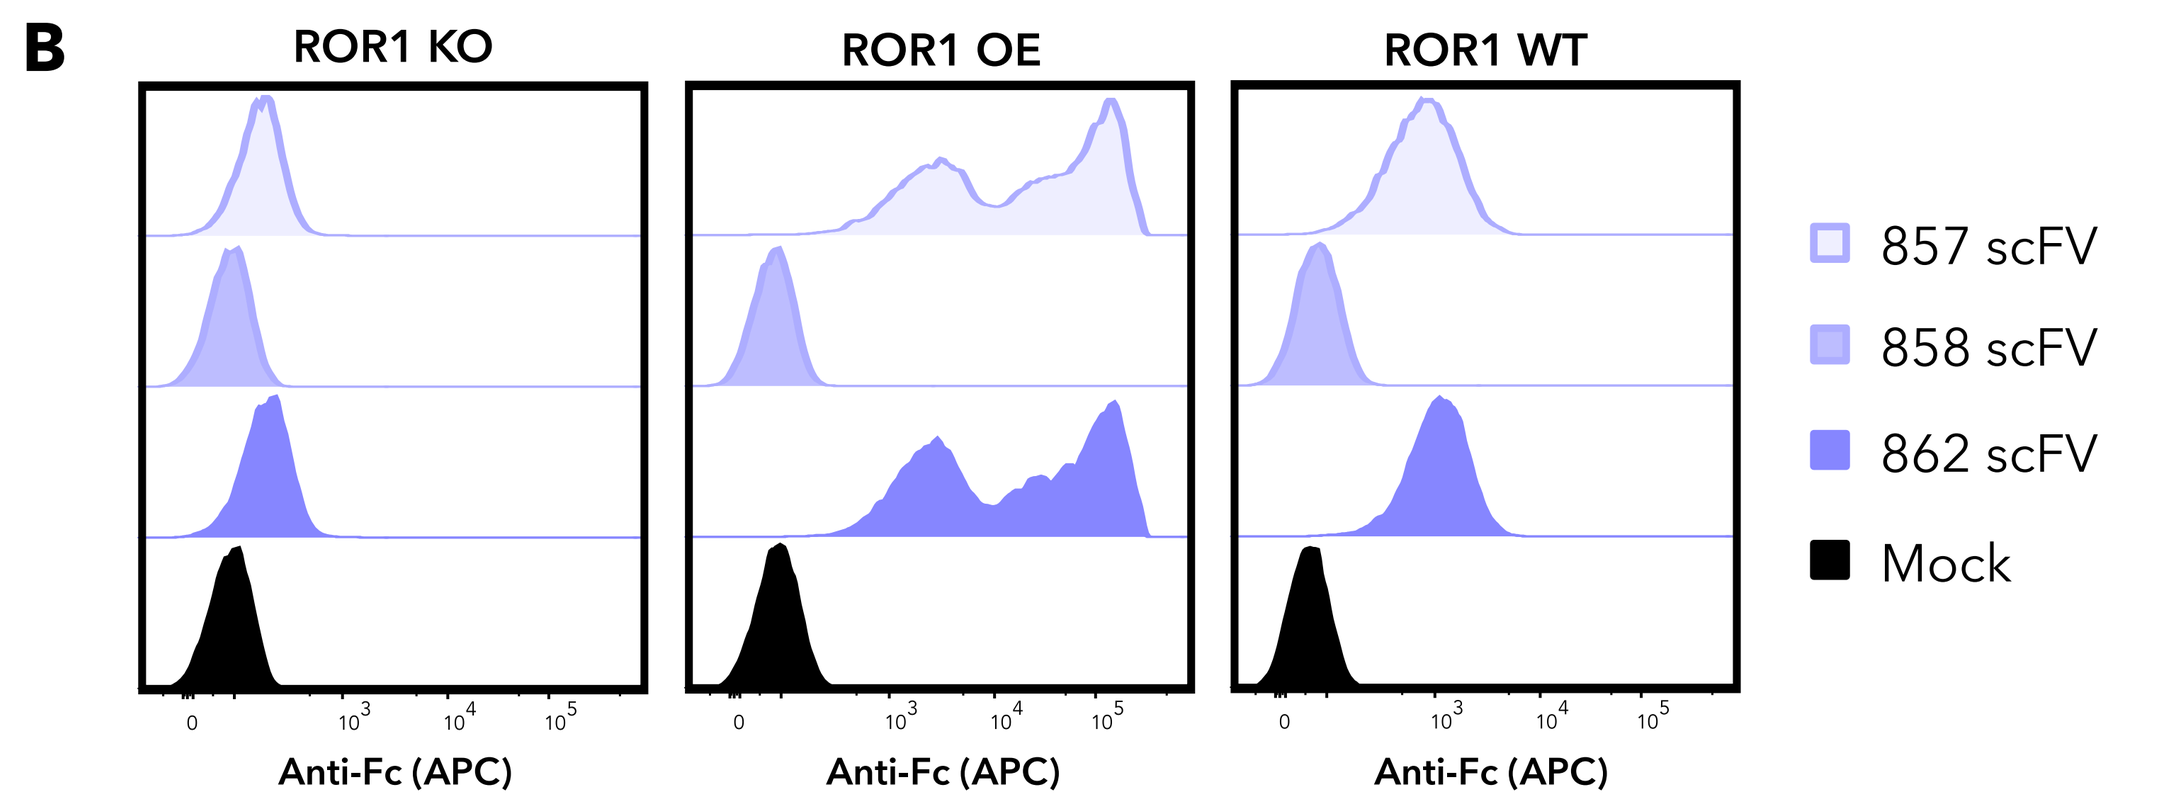
B
ROR1 KO
ROR1 OE
ROR1 WT
Anti-Fc (APC)
857 scFV
858 scFV
862 scFV
Mock

Fig 4. ROR1 CAR constructs evaluated in CAR-iNKs (**A**) Anti-ROR1 CAR constructs pCB7140, pCB7141, and pCB7142 containing the novel scFVs 857, 858, and 862 respectively. (**B**) Soluble ROR1 scFV-Fc fusion proteins bind to ROR1 on SKOV-3 tumor cells with high affinity and specificity. Supernatants containing 857, 858, and 862 scFv-Fc fusion proteins harvested from transiently transfected HEK293T cells were incubated with ROR1 knockout (KO), ROR1 over-expressing (OE), and ROR1 wild-type (WT) SKOV-3 tumor cells followed by labelling with anti-human Fc-APC conjugated secondary. Mock = supernatant from HEK293T cells transfected with no plasmid.

ROR1 tumor target and CAR construct (*cont'd*)

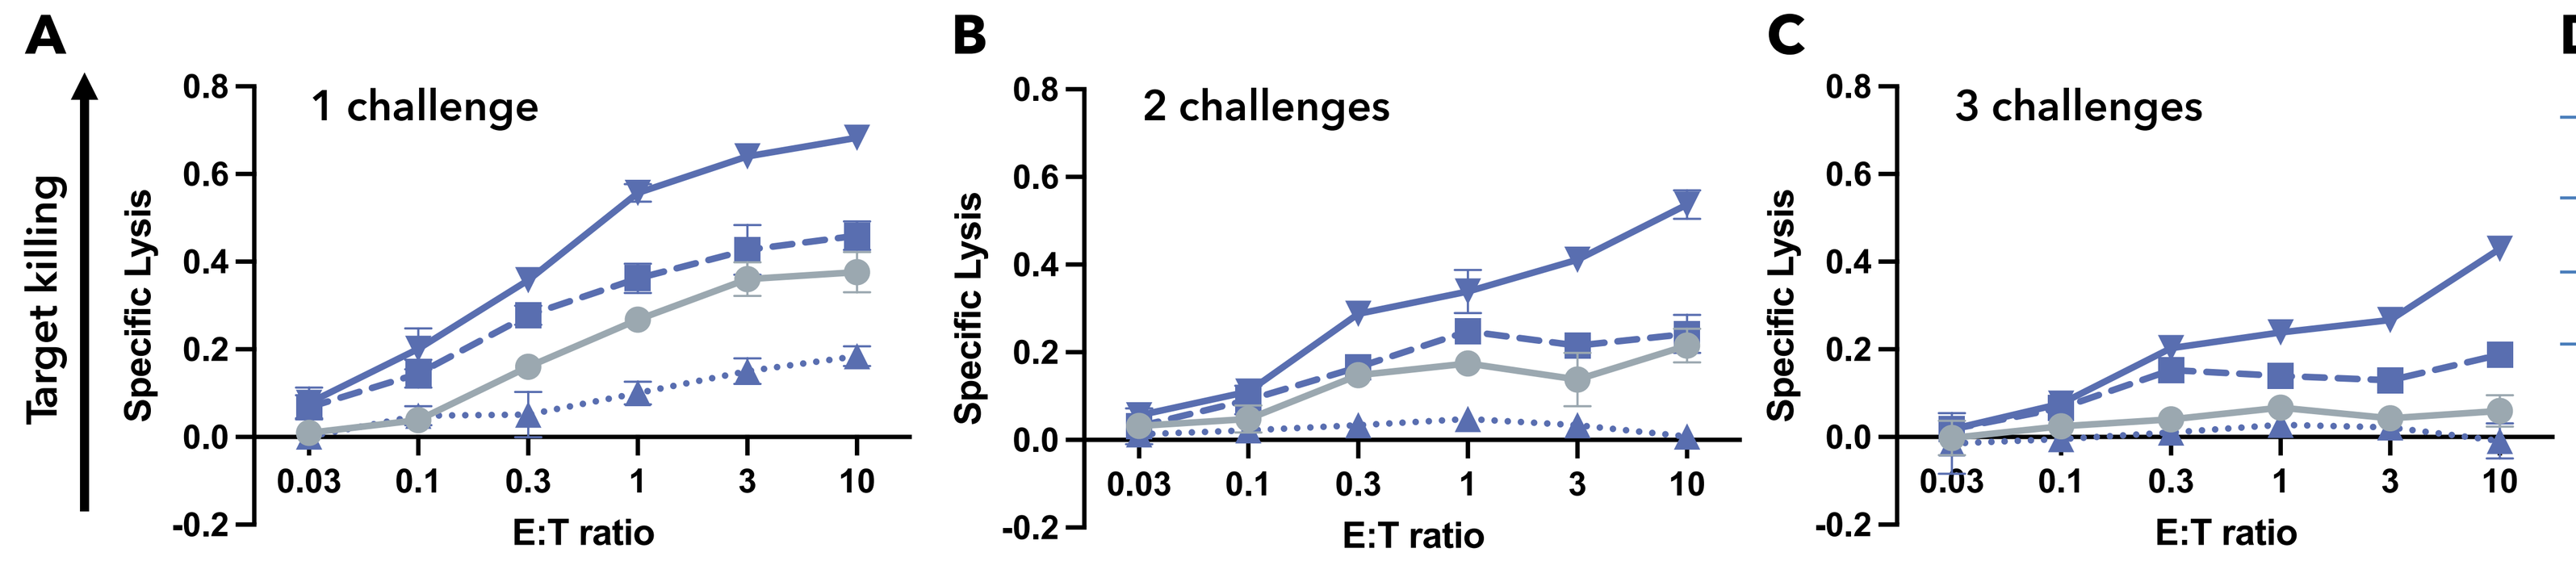
A
1 challenge
B
2 challenges
C
3 challenges
Target killing
Specific Lysis
E:T ratio

D

	% CAR+
WT iNKs	0.32%
pCB7140 CAR-iNKs	96.99%
pCB7141 CAR-iNKs	92.46%
pCB7142 CAR-iNKs	92.17%

Fig 5. CAR-iNKs in serial killing activity assays against JeKo-1 tumor target cell line. pCB7142 CAR-iNKs demonstrate improved cytotoxicity against JeKo-1 tumor model in serial killing assay compared to pCB7140 and pCB7142. CAR-iNKs co-cultured with JeKo-1 target tumor cells at a range of effector-to-target (E:T) ratios for 4 hours (**A**) received no prior challenges, (**B**) received one prior challenge, and (**C**) received 2 prior challenges, prior challenges at 2:1 E:T ratio, with 24 hrs between each challenge. CAR-iNKs normalized to count of CD56+ cells prior to initiation of final challenge. Target JeKo-1 cells pre-stained with cell trace violet (CTV) after 4-hour co-culture all wells stained with PI. Live targets (LT) determined by CTV positive, PI negative counts. Specific lysis calculated as follows: 1- LT (test condition) / LT (target only). (**D**) Percentage of CAR+ NK cells in the populations used for serial killing assay.

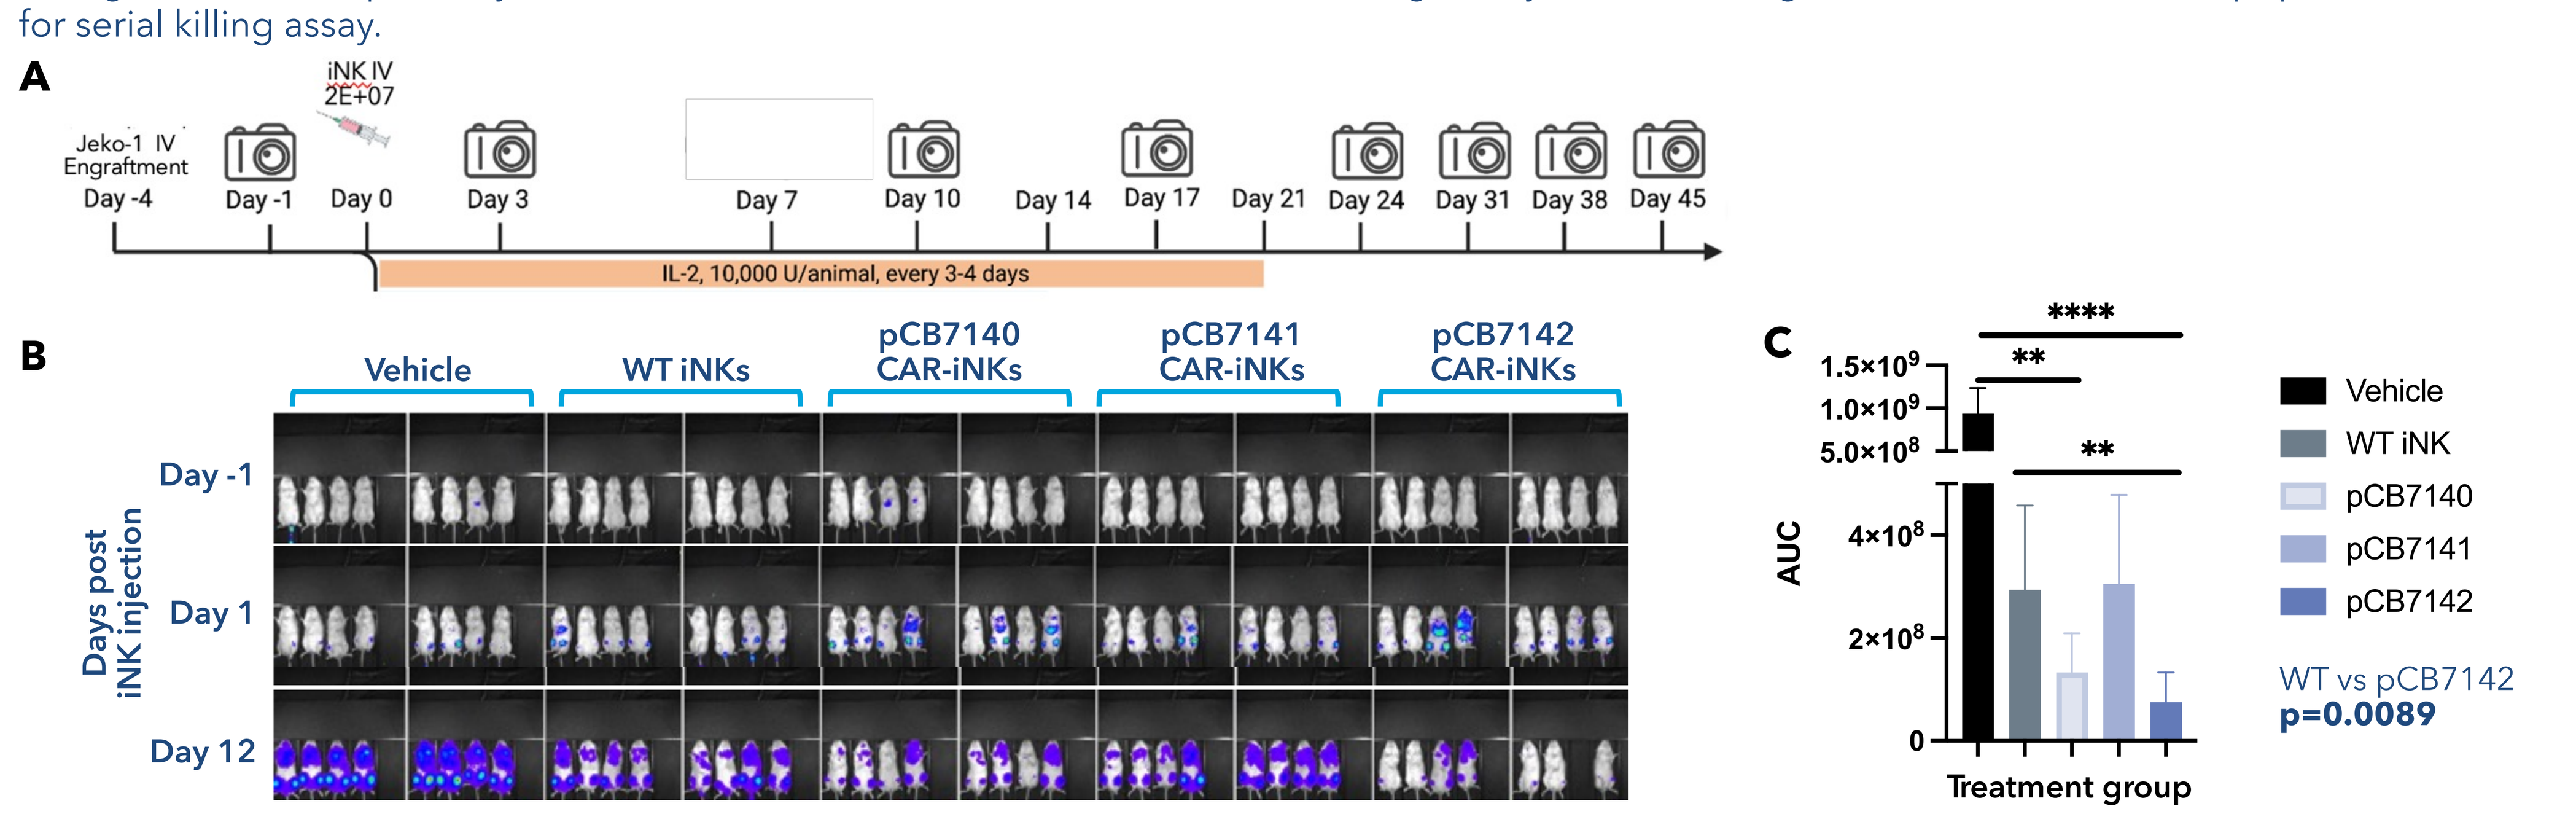
A
Jeko-1 IV Engraftment
iNK IV 2E+07
Day -4
Day -1
Day 0
Day 3
Day 7
Day 10
Day 14
Day 17
Day 21
Day 24
Day 31
Day 38
Day 45
IL-2, 10,000 U/animal, every 3-4 days
B
Vehicle
WT iNKs
pCB7140 CAR-iNKs
pCB7141 CAR-iNKs
pCB7142 CAR-iNKs
Days post iNK injection
Day -1
Day 1
Day 12
C
AUC
Treatment group
Vehicle
WT iNK
pCB7140
pCB7141
pCB7142
WT vs pCB7142
p=0.0089

Fig 6. pCB7142 CAR-iNKs demonstrate significant reduction in tumor burden compared to WT iNKs in JeKo-1 tumor-engrafted hIL15 TG-NOG mice
(**A**) Schematic of *in vivo* study design. 5E+05 JeKo-1-GFPluc tumor cells injected IV in hIL15-TG-NOG mice at day -4. Tumor engrafted animals were IVIS imaged on day -1 and randomized across treatment groups. 2E+07 iNKs were engrafted IV on day 0 and imaged by IVIS twice weekly.
(**B**) Bioluminescent imaging of tumor burden and localization in vehicle control and iNK treated animals at day 1 and day 12 post iNK engraftment.
(**C**) AUC analysis of bioluminescence intensity for vehicle and treatment groups from day -1 to 12. P-value annotations ** <0.01, **** <0.0001.

ROR1 antigen expression on tumor cell lines

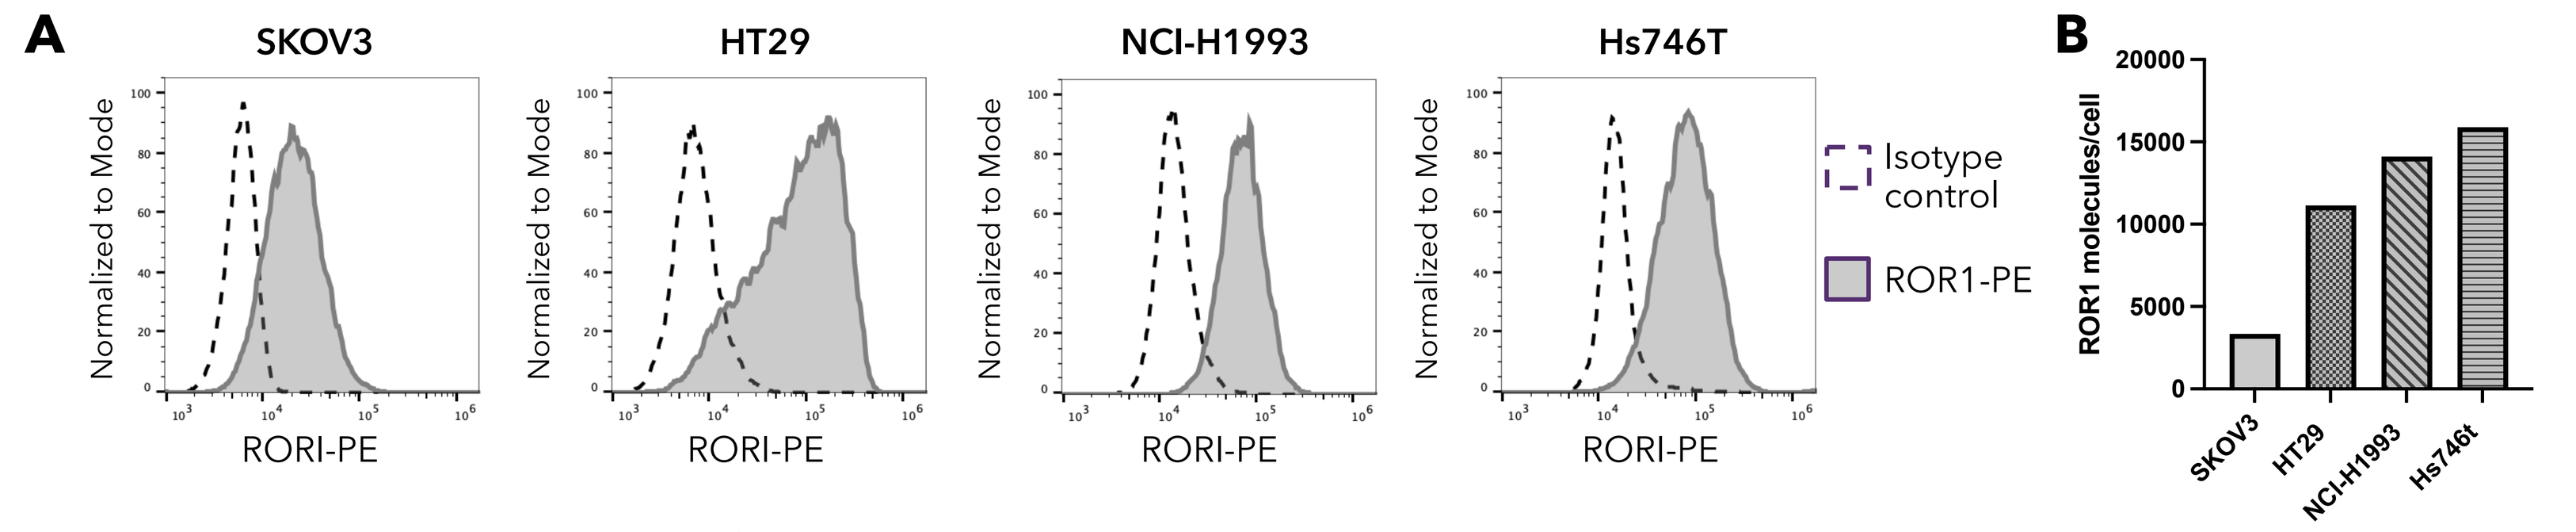
A
SKOV3
HT29
NCI-H1993
Hs746T
Normalized to Mode
RORI-PE
Isotype control
ROR1-PE
B
ROR1 molecules/cell
SKOV3
HT29
NCI-H1993
Hs746t

Fig 7. ROR1 antigen expression on solid tumor cell lines.
(**A**) ROR1 expression profile on SKOV3 (ovarian), HT29 (colorectal), NCI-H1993 (lung), and Hs746t (gastric) tumor cell lines. (**B**) ROR1 antigen density quantification on solid tumor cell lines using BD Quantibrite™ beads.

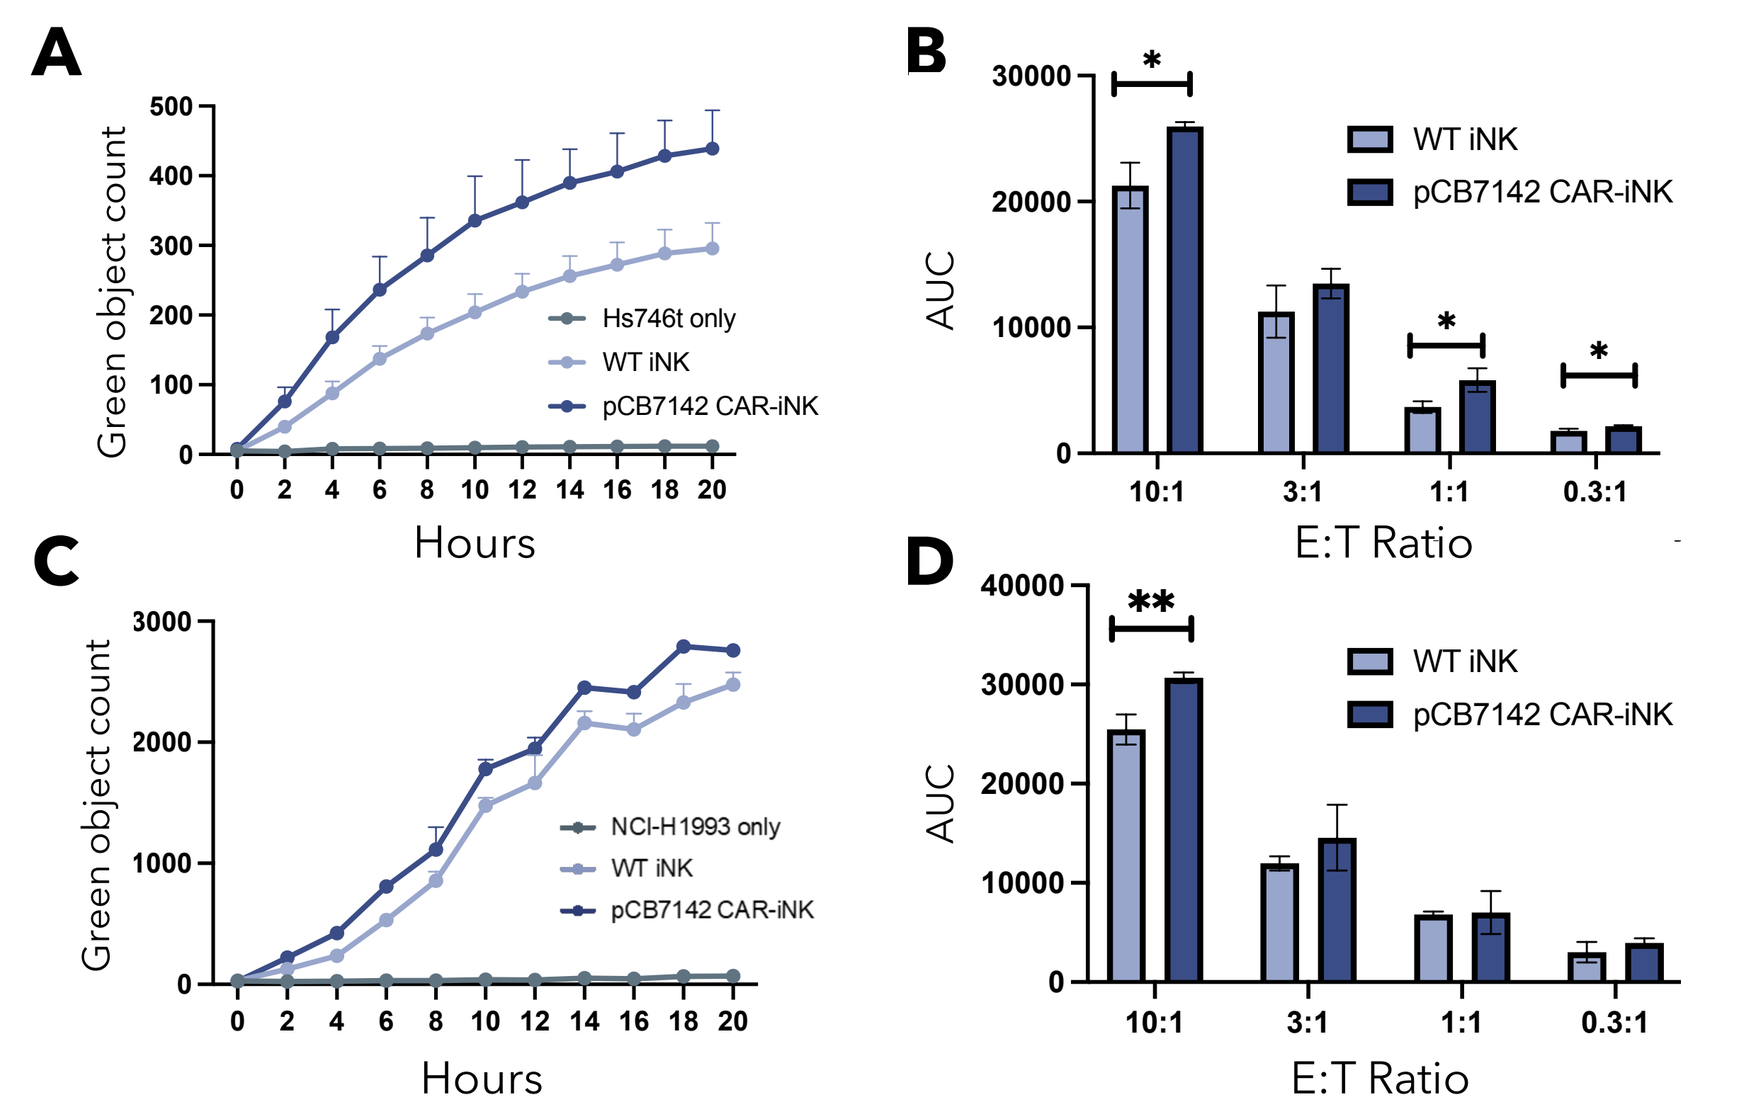
A
Green object count
Hours
Hs746t only
WT iNK
pCB7142 CAR-iNK
B
AUC
E:T Ratio
WT iNK
pCB7142 CAR-iNK
C
NCI-H1993 only
WT iNK
pCB7142 CAR-iNK
D

Fig 8. Increased cytotoxicity of CAR-iNKs against Hs746t gastric tumor and NCI-H1993 lung tumor cell lines.
(**A**) Incucyte-based caspase 3/7 assay of WT iNKs and pCB7142 CAR-iNKs against Hs746t at 1:1 E:T ratio. Green object count representative of apoptotic cells labelled with caspase 3/7 cleavage green dye. Increased green object count indicative of increased cytotoxicity of effector cells. (**B**) AUC of green object count from caspase 3/7 assay at 10:1, 3:1, 1:1, and 0.3:1 E:T ratios. P-value annotation * <0.05.
(**C**) Incucyte-based caspase 3/7 assay of WT iNKs and pCB7142 CAR-iNKs against NCI-H1993 at 10:1 E:T ratio. Green object count representative of apoptotic cells labelled with caspase 3/7 cleavage green dye. Increased green object count indicative of increased cytotoxicity of effector cells.
(**D**) AUC of green object count from caspase 3/7 assay at 10:1, 3:1, 1:1, and 0.3:1 E:T ratios. P-value annotation ** <0.01.

iNK armoring strategies

1. CBLB knockout (CBLB: E3 ubiquitin ligase that regulates NK receptor expression)

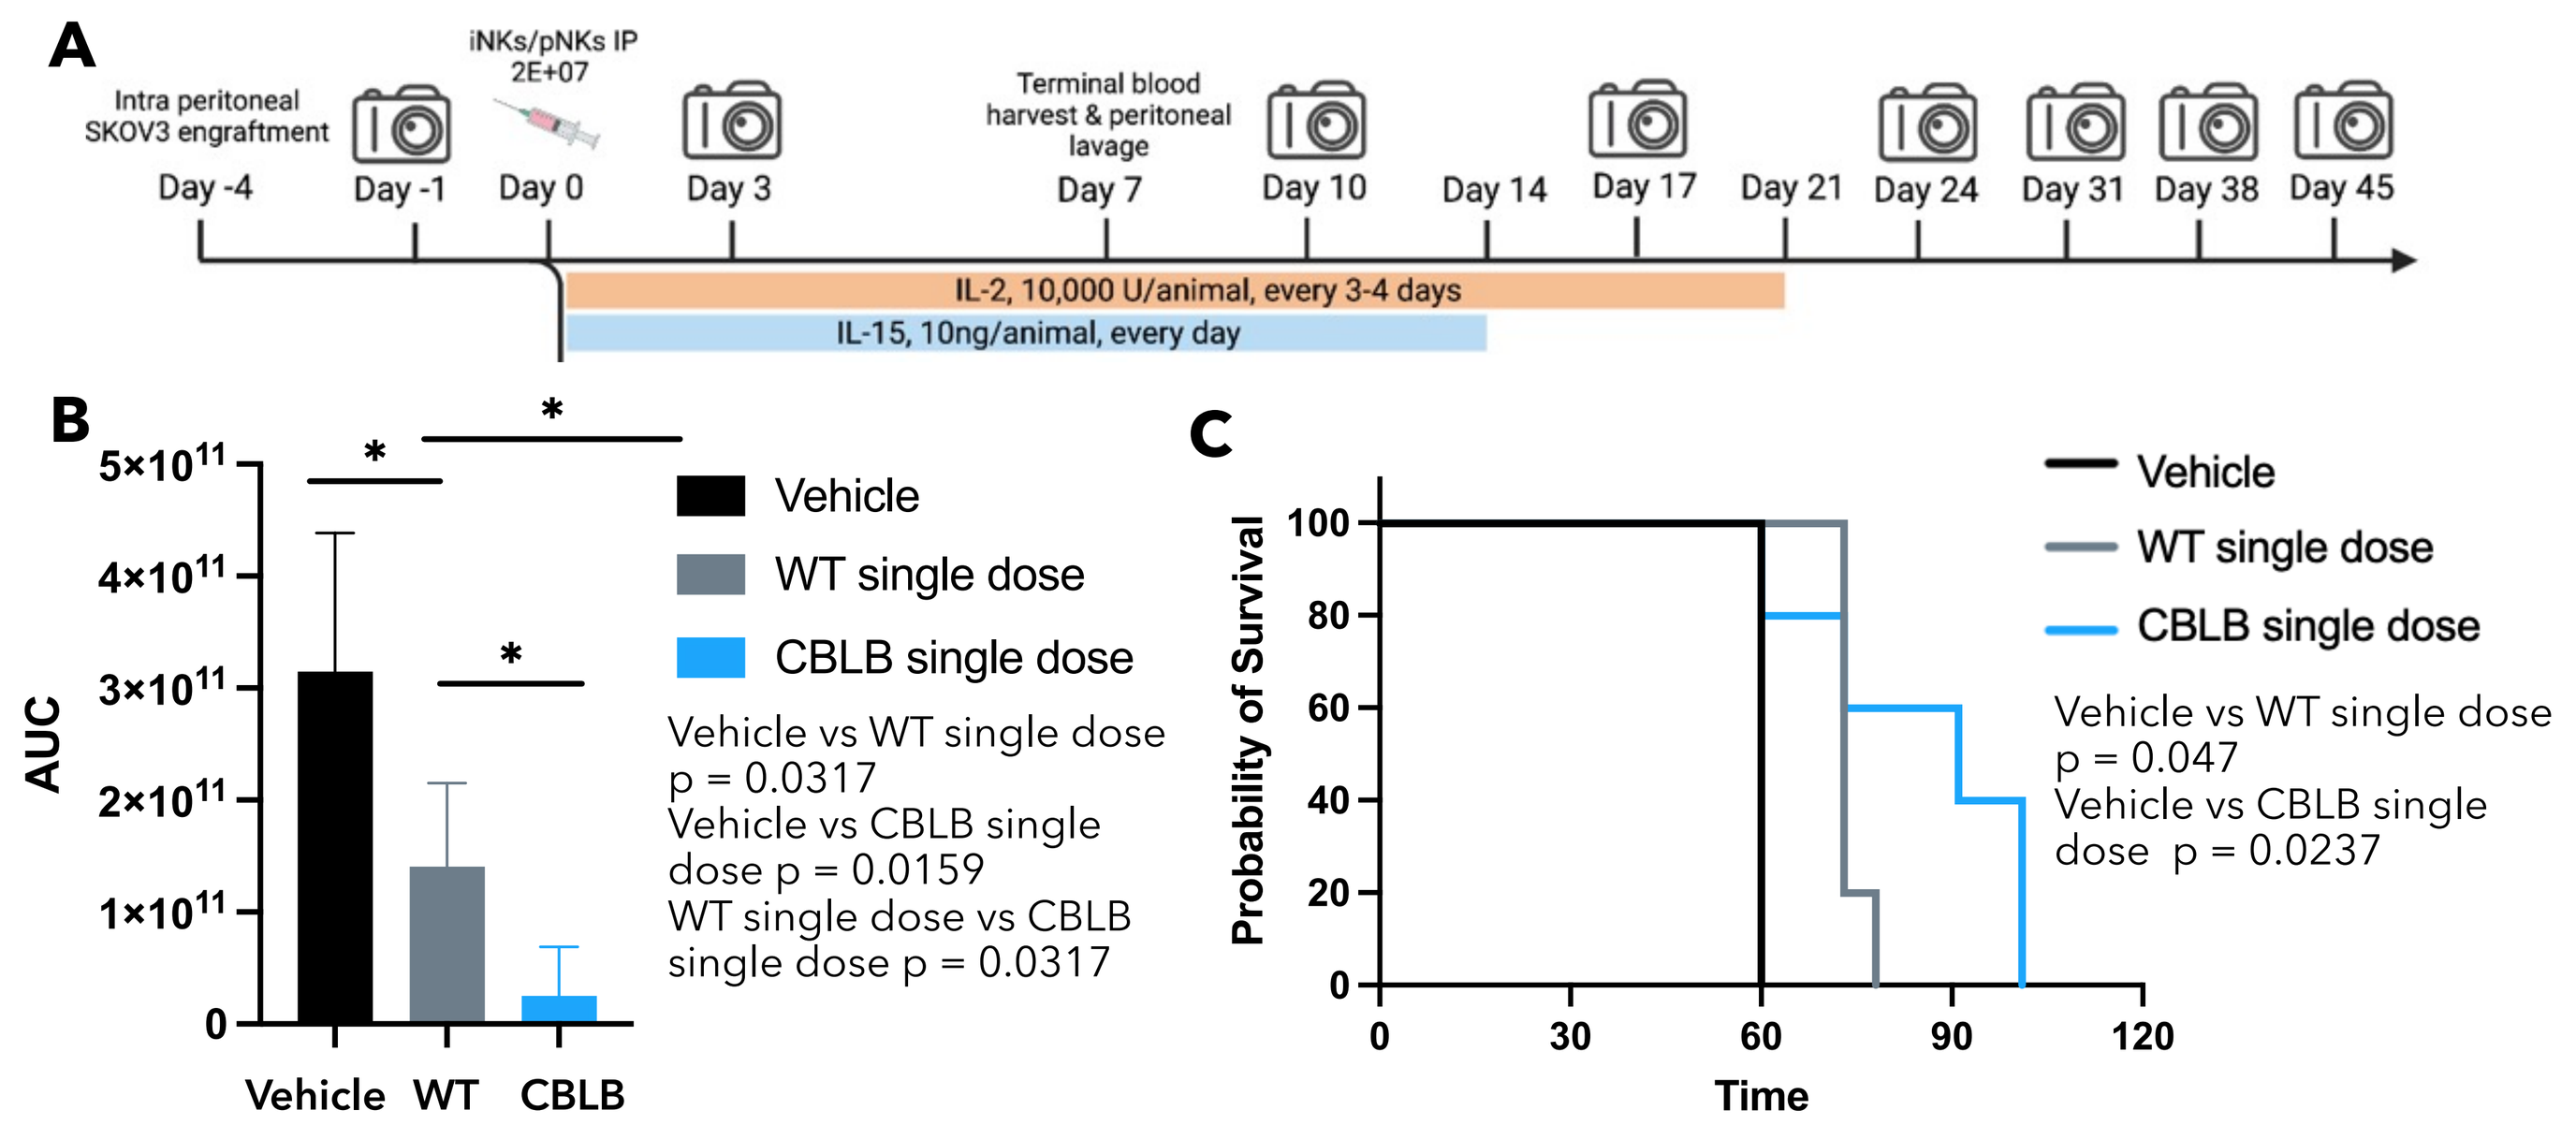
A
Intra peritoneal SKOV3 engraftment
iNKs/pNKs IP 2E+07
Terminal blood harvest & peritoneal lavage
IL-2, 10,000 U/animal, every 3-4 days
IL-15, 10ng/animal, every day
B
AUC
Vehicle
WT single dose
CBLB single dose
Vehicle vs WT single dose p = 0.0317
Vehicle vs CBLB single dose p = 0.0159
WT single dose vs CBLB single dose p = 0.0317
C
Probability of Survival
Time
Vehicle vs WT single dose p = 0.047
Vehicle vs CBLB single dose p = 0.0237

Fig 9. CBLB KO iNKs exhibit significant enhancement in antitumor activity compared to WT iNKs in SKOV3-engrafted NSG model. (**A**) Schematic of *in vivo* study design. 3E+05 SKOV3-GFPluc tumor cells injected IP in NSG animals at day -4. Tumor-engrafted animals were IVIS imaged on day -1 and randomized across treatment groups. 2E+07 iNKs were engrafted IP on day 0 and imaged by IVIS twice weekly. (**B**) AUC analysis of bioluminescence intensity for vehicle and treatment groups from day -1 to 45 (AUC analysis only for duration when all vehicle control animals remaining). P-value annotations * < 0.05. (**C**) Probability of survival for vehicle and iNK treatment groups.

2. B2M knockout and B2M-HLA-E knock-in (Class I expression disruption/engineering)

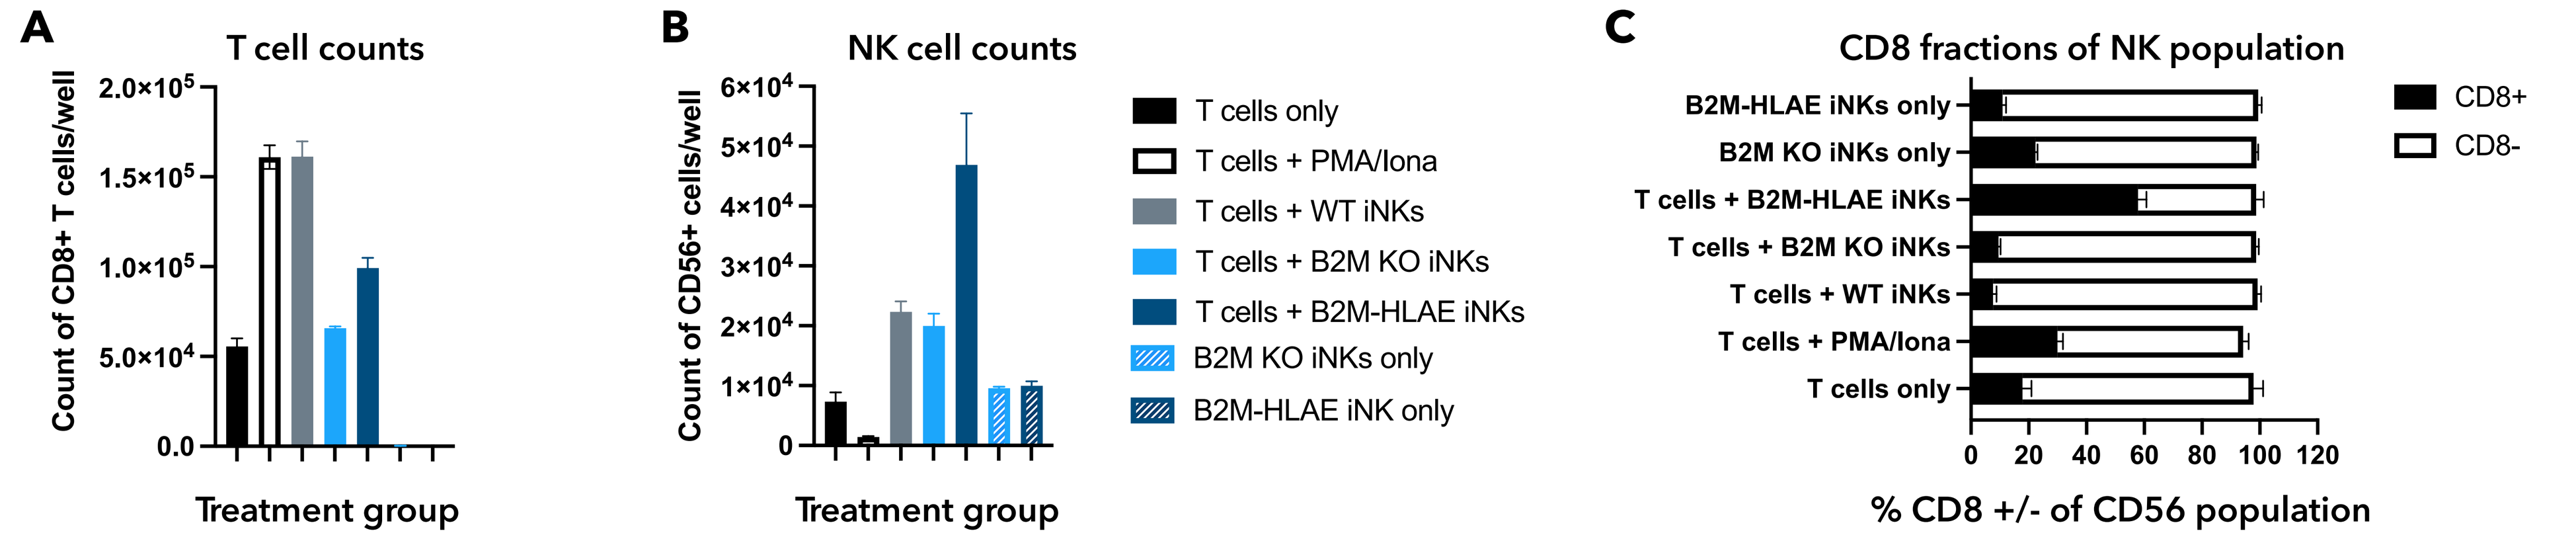
A
T cell counts
Count of CD8+ T cells/well
Treatment group
B
NK cell counts
Count of CD56+ cells/well
T cells only
T cells + PMA/Iona
T cells + WT iNKs
T cells + B2M KO iNKs
T cells + B2M-HLAE iNKs
B2M KO iNKs only
B2M-HLAE iNK only
C
CD8 fractions of NK population
B2M-HLAE iNKs only
B2M KO iNKs only
T cells + B2M-HLAE iNKs
T cells + B2M KO iNKs
T cells + WT iNKs
T cells + PMA/Iona
T cells only
CD8+
CD8-
% CD8 +/- of CD56 population

Fig 10. The B2M knockout/B2M-HLA-E knock-in reduces T cell expansion and promotes iNK cell expansion. CD8+ T cells enriched by negative selection from PBMCs were co-cultured with WT iNKs, B2M KO iNKs, and B2M-HLA-E iNKs at 4:1 (T cells : NK cells) for 6 days. (**A**) T cell expansion evaluated at day 6 by CD8 staining with counting beads, indicating expansion of T cells under WT iNK co-culture and positive control PMA/ ionamycin treatment, minimal T cell expansion observed in B2M KO and B2M-HLA-E iNK co-culture conditions. (**B**) iNK counts at day 6 were evaluated by CD56 staining and counting beads. Relative to control iNK only conditions, no T cell-mediated iNK killing was observed. B2M-HLA-E iNKs expanded under T cell co-culture conditions. (**C**) Increased predominance of a CD56+CD8+ population in B2M-HLA-E iNK cultured with T cells was observed.

3. Membrane-bound IL-15/IL15RA fusion knock-in (Tethered cytokine support for enhanced function)

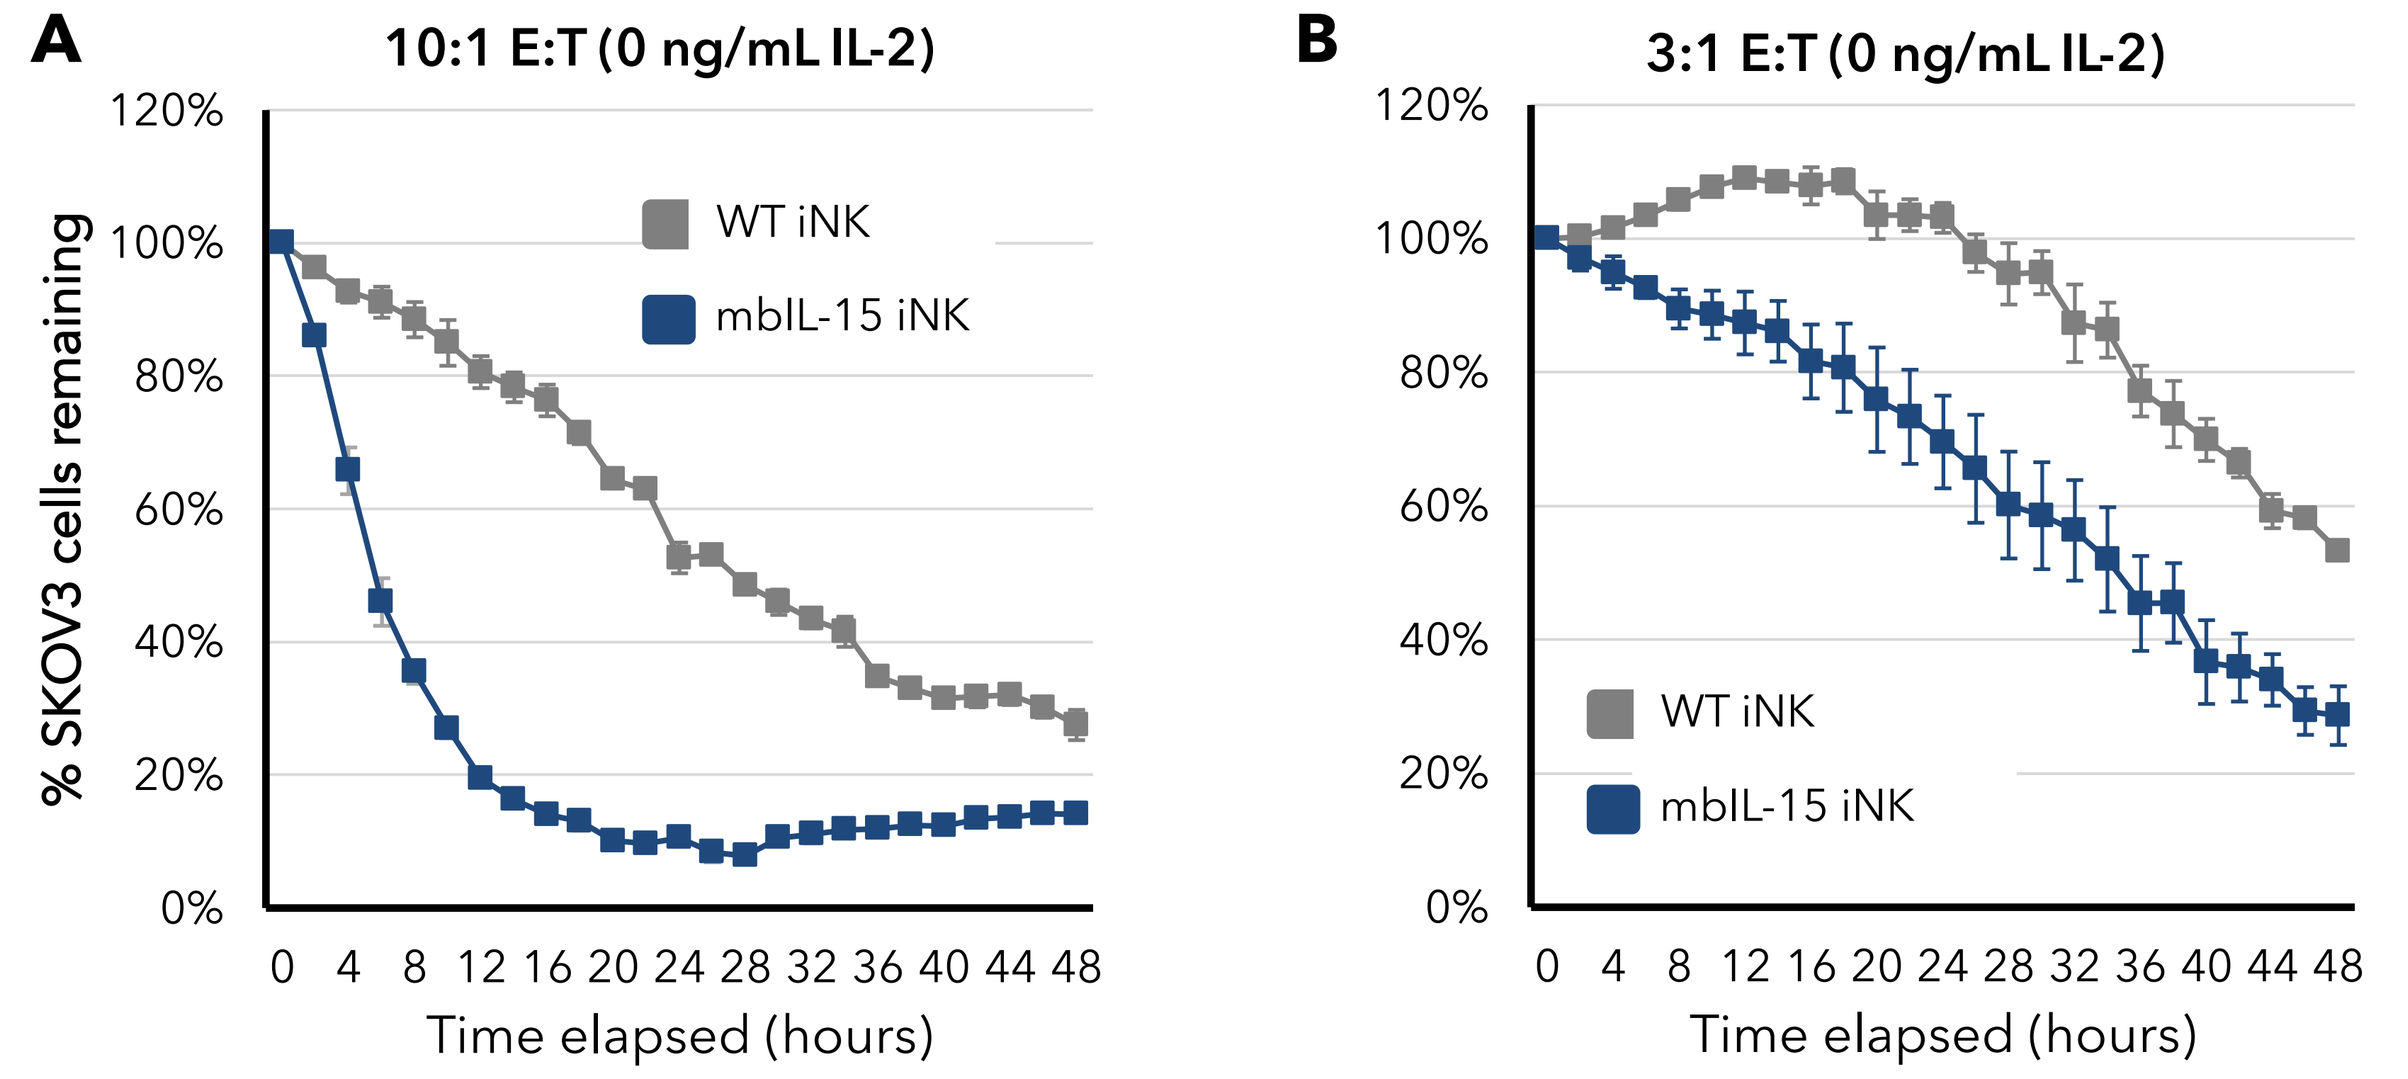
A
10:1 E:T (0 ng/mL IL-2)
% SKOV3 cells remaining
WT iNK
mbIL-15 iNK
Time elapsed (hours)
B
3:1 E:T (0 ng/mL IL-2)

Fig 11. Membrane-bound IL-15/IL-15RA fusion (mbIL-15) engineered iNKs demonstrate high cytotoxicity against SKOV3 ovarian tumor cell line in the absence of exogenous cytokine support. mbIL-15-engineered iNKs at an effector-to-target ratio of 10:1 (**A**) and 3:1 (**B**) in the absence of exogenous cytokines. SKOV-3 GFP line in combination with YO-PRO-3 viability dye was used to measure viable SKOV-3 cells.

Conclusions

- We have developed a robust and reproducible platform for differentiating and expanding genome-edited iPSCs into CAR-iNK cells targeting the ROR1 tumor antigen
- We have demonstrated three armoring strategies for our CAR-iNK platform that we are evaluating for use in CB-020:
 1. *CBLB* knockout **2.** *B2M* knockout and B2M-HLA-E knock-in **3.** Membrane-bound IL-15/IL-15RA fusion knock-in
- iNK cells innately exhibit potent antitumor activity against solid tumors and armoring them with an anti-ROR1 CAR and knockout/knock-in of regulatory genes is designed to augment their trafficking, persistence, specificity, and antitumor activity against ROR1⁺ solid tumors

© 2022 Caribou Biosciences, Inc.
"Caribou Biosciences" and Caribou's logo are registered trademarks of Caribou Biosciences, Inc.

CORRESPONDING AUTHOR: Rodolfo Gonzalez (rudy.gonzalez@cariboubio.com)
Caribou Biosciences, Inc.

12th AACR-JCA Joint Conference: Breakthroughs in Cancer Research – Translating Knowledge into Practice
December 10-14, 2022 – Maui, Hawaii