POSTER # 309

CARIBOU BIOSCIENCES®

Improved genome editing with Cas12a and Cas9 chRDNA platform in T, NK, B, and iPS cells

Paul Donohoue[1], Martin Pacesa[2], Elaine Lau[1‡], Bastien Vidal[1‡], Matthew J. Irby[1‡], David B. Nyer[1‡], Tomer Rotstein[1‡], Mikey Loncaric[1], Emma Ciminelli[1], Erica Vander Mause[1], Tony Muñoz[1], Lynda Banh[1‡], Mckenzi S. Toh[1‡], Glen Churchward[1], Rachel Gonzalez[1], Jason Gibson[1‡], Bryan Kohrs[1], Kevin Baek[1‡], Arthur L.G. Owen[1], Euan M. Slorach[1‡], Megan van Overbeek[1‡], Andrew P. May[1‡], Elizabeth Garner[1], Martin Jinek[2], Peter Cameron[1‡]

[1]Caribou Biosciences, Inc., Berkeley, CA 94710, United States; [2]Department of Biochemistry, University of Zurich, CH-8057 Zurich, Switzerland

‡Former Caribou employee

Abstract

CRISPR-based genome editing of primary human T cells has the potential to revolutionize cell therapies. However, substantial improvements in CRISPR-Cas9 specificity are needed to significantly reduce its off-target activity in cells. Here we show that CRISPR hybrid RNA-DNA (chRDNA) guides designed with both RNA and DNA nucleotides are a highly effective approach to increase the specificity of Cas9 and Cas12a genome editors while preserving on-target editing activity. Across multiple genomic targets in primary human T cells, we show that 2'-deoxynucleotide positioning affects guide activity in a sequence-dependent manner, and we leveraged this observation to engineer chRDNA guides with minimal to no detectable off-target activities. To further enhance the capability of the Cas12a chRDNA platform for engineering of cell therapies, we optimized the nuclear trafficking sequence, thereby improving editing efficiency even at low concentrations of Cas12a across multiple primary cell types.

Cas9 chRDNA technology demonstrates improved specificity compared to an engineered Cas9 variant

The initial investigation of DNA incorporation into Cas9 RNA guides showed that Cas9 tolerated high levels, as high as 60%, of RNA replacement in the target-complimentary spacer region. To identify ideal chRDNA designs, we employed an iterative approach to empirically determine the position and combination of DNA bases through successive rounds of testing to reduce editing at off-target ("off") sites while maintaining on-target ("on") editing rates comparable to all-RNA guides in human T cells (A). To understand the biophysical impact of chRDNA guides we co-crystalized Cas9 and corresponding on-target DNAs bound to either all-RNA crRNA (PDB:7OX9) or chRDNA guides (PDB:7OX8). We determined that DNA incorporation drove a conformation deformation of the target-spacer heteroduplex resulting in a "kinking" of the duplex and displacement of key protein domains in the Cas9 nuclease critical for initiation of cleavage (B). We then compared optimized chRDNAs for a series of targets to a commercially available all-RNA crRNA guide and an engineered high-fidelity ("Hifi") Cas9 variant. While the HifiCas9 showed improved specificity compared to wildtype ("wt") Cas9 with all-RNA guides, optimized chRDNAs fully prevented off-target editing at the target tested (C).

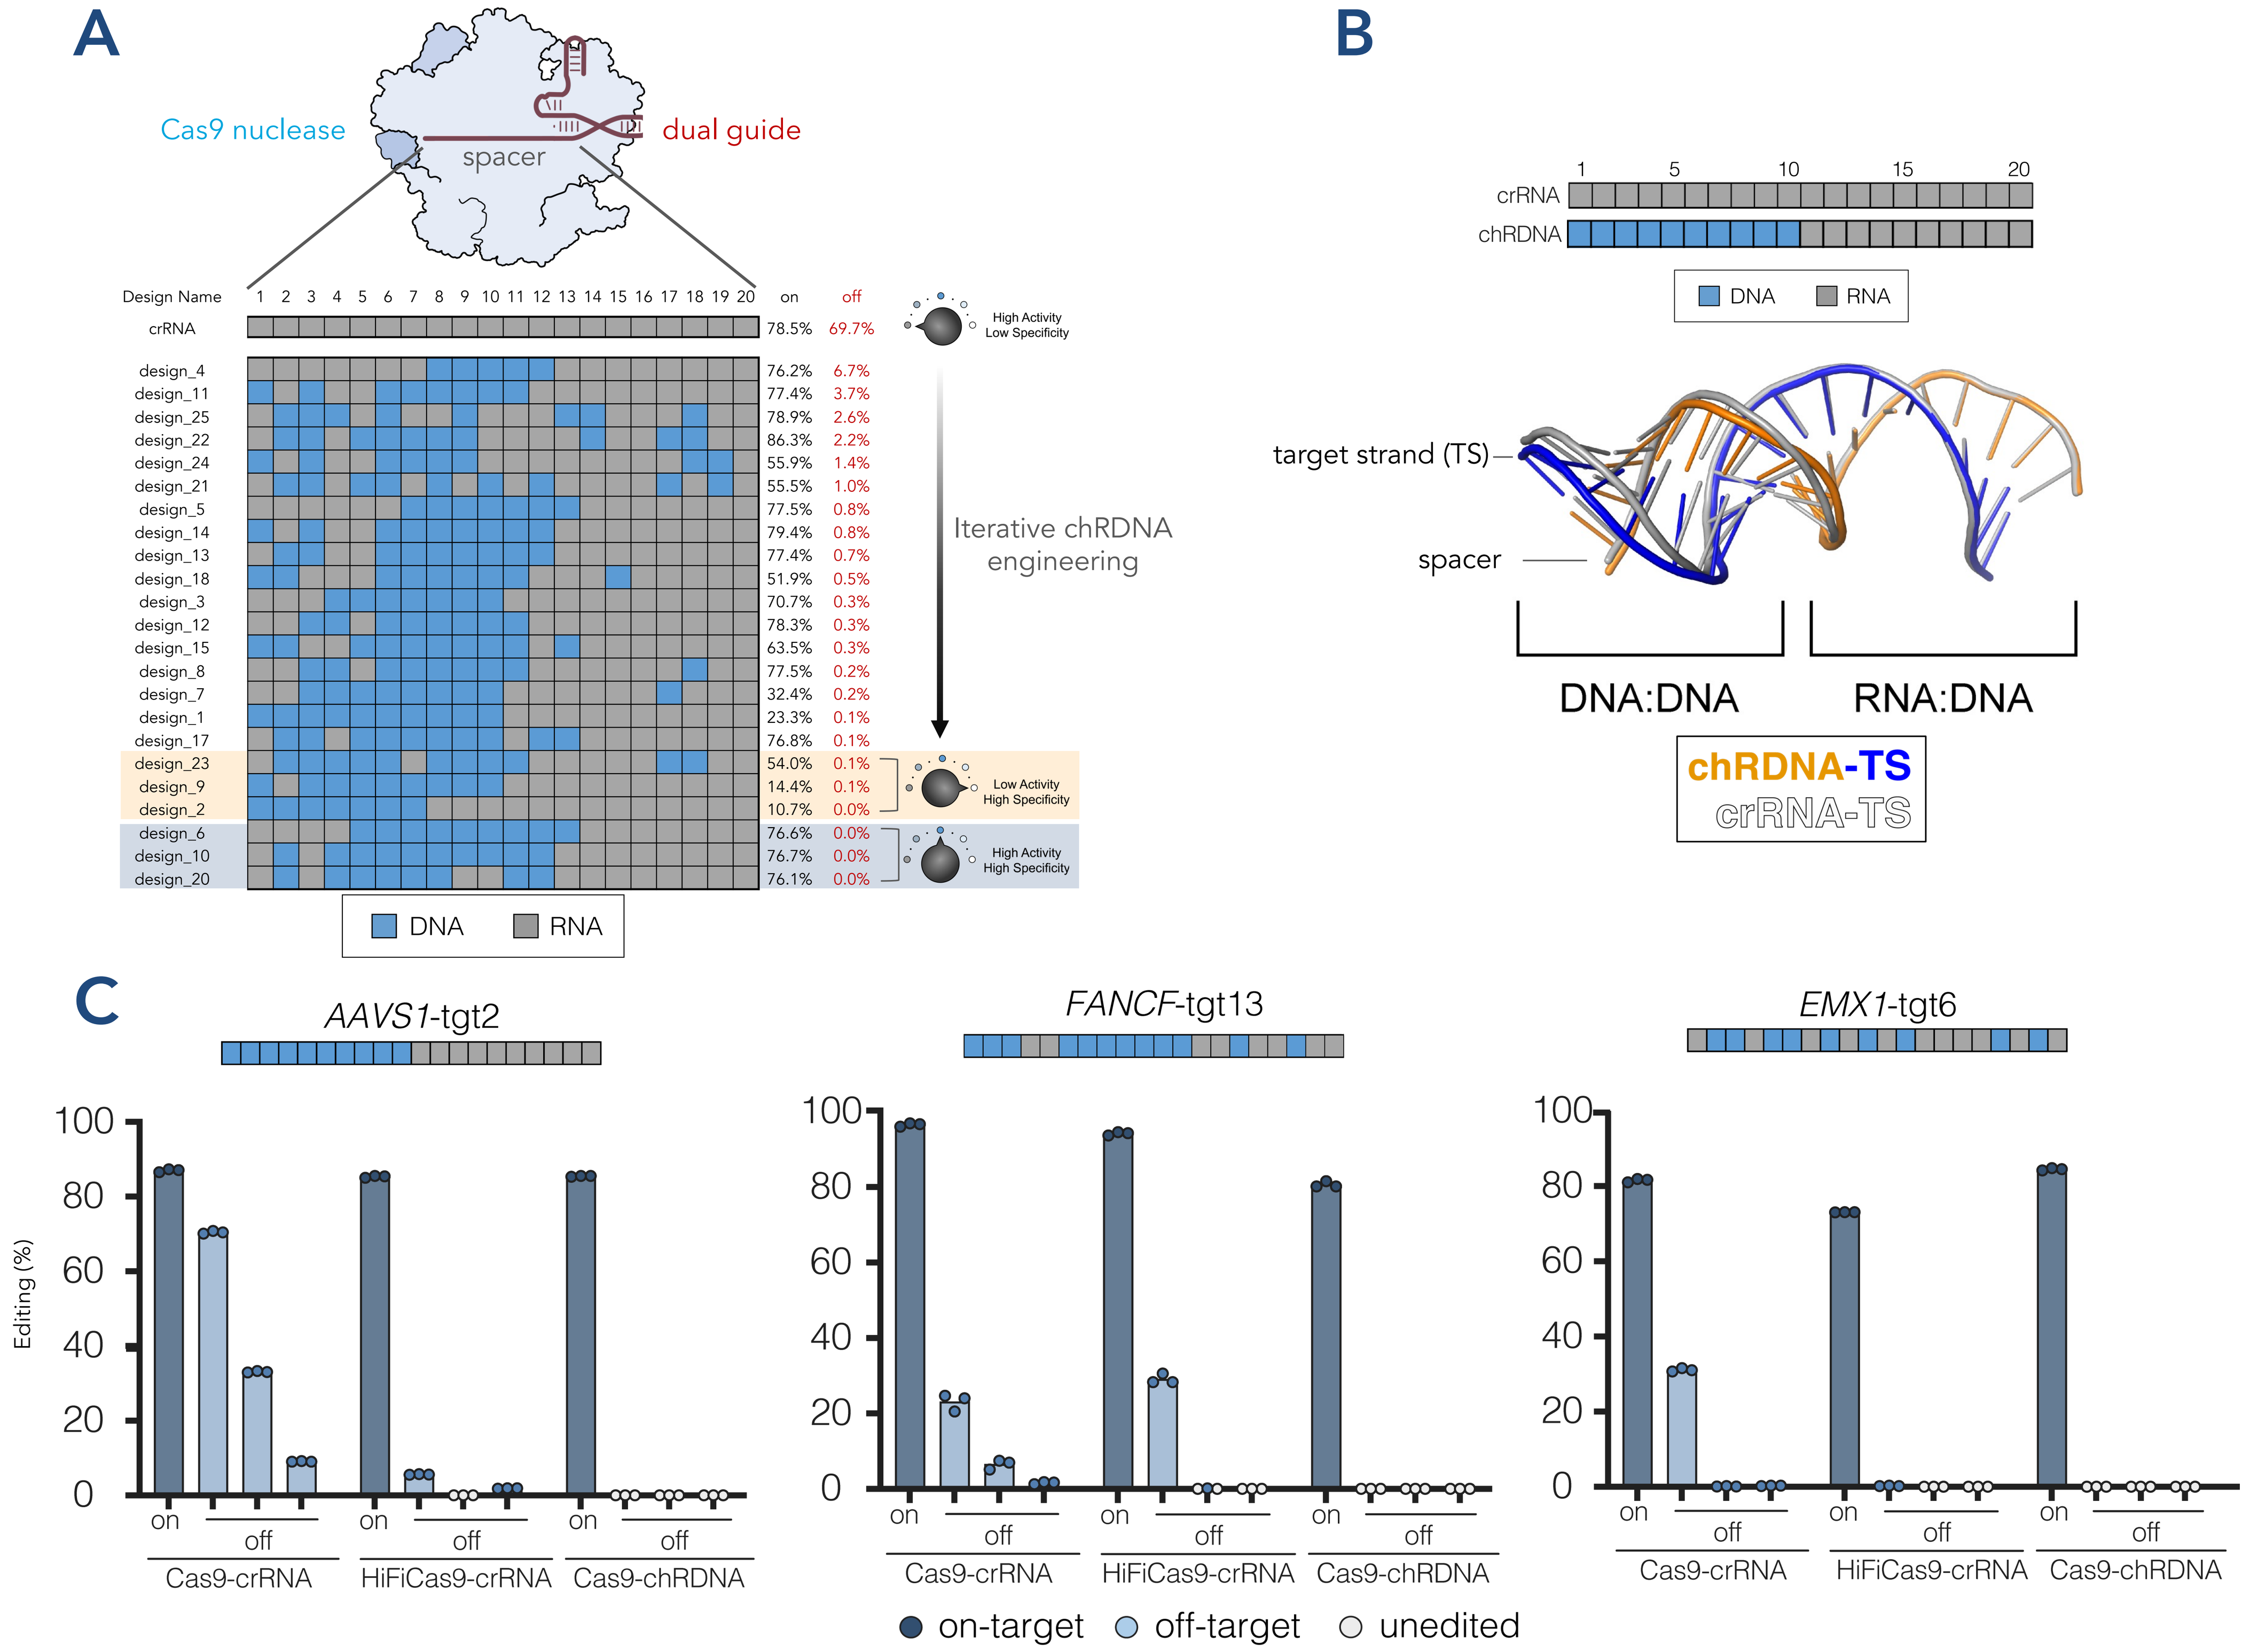
A
Cas9 nuclease
spacer
dual guide
Design Name
on
off
crRNA 78.5% 69.7%
High Activity Low Specificity
Iterative chRDNA engineering
Low Activity High Specificity
High Activity High Specificity
DNA RNA
B
crRNA
chRDNA
DNA RNA
target strand (TS)
spacer
DNA:DNA
RNA:DNA
chRDNA-TS
crRNA-TS
C
AAVS1-tgt2
FANCF-tgt13
EMX1-tgt6
Editing (%)
on
off
Cas9-crRNA
HiFiCas9-crRNA
Cas9-chRDNA
on-target off-target unedited

chRDNA guides can improve the specificity of the Cas12a nuclease system

In contrast to Cas9, initial chRDNA designs with the Cas12a nuclease showed poor tolerance for abundant DNA load in the guide spacer region. To identify DNA-amenable positions, we developed a new screening pipeline where we tested twenty chRDNAs with a single DNA base at each position in the spacer region (A). We then blended individual DNA-permissive positions to generate new designs with multiple DNA bases positioned in the spacer without compromising on-target editing (B). Final chRNDA designs were tested for on- and off-target editing efficiencies in human T cells (C). Despite Cas12a all-RNA guides generally showing low off-target editing (compared to Cas9 all-RNA guides), optimized chRDNA guides further improved specificity resulting in no off-target edits above 0.1% when analyzed by next-generation sequencing. These results demonstrate the utility of the chRDNA platform across different classes of Cas nucleases and that incorporation of DNA bases into the spacer region sensitizes Cas nucleases to stable engagement at off-target sites.

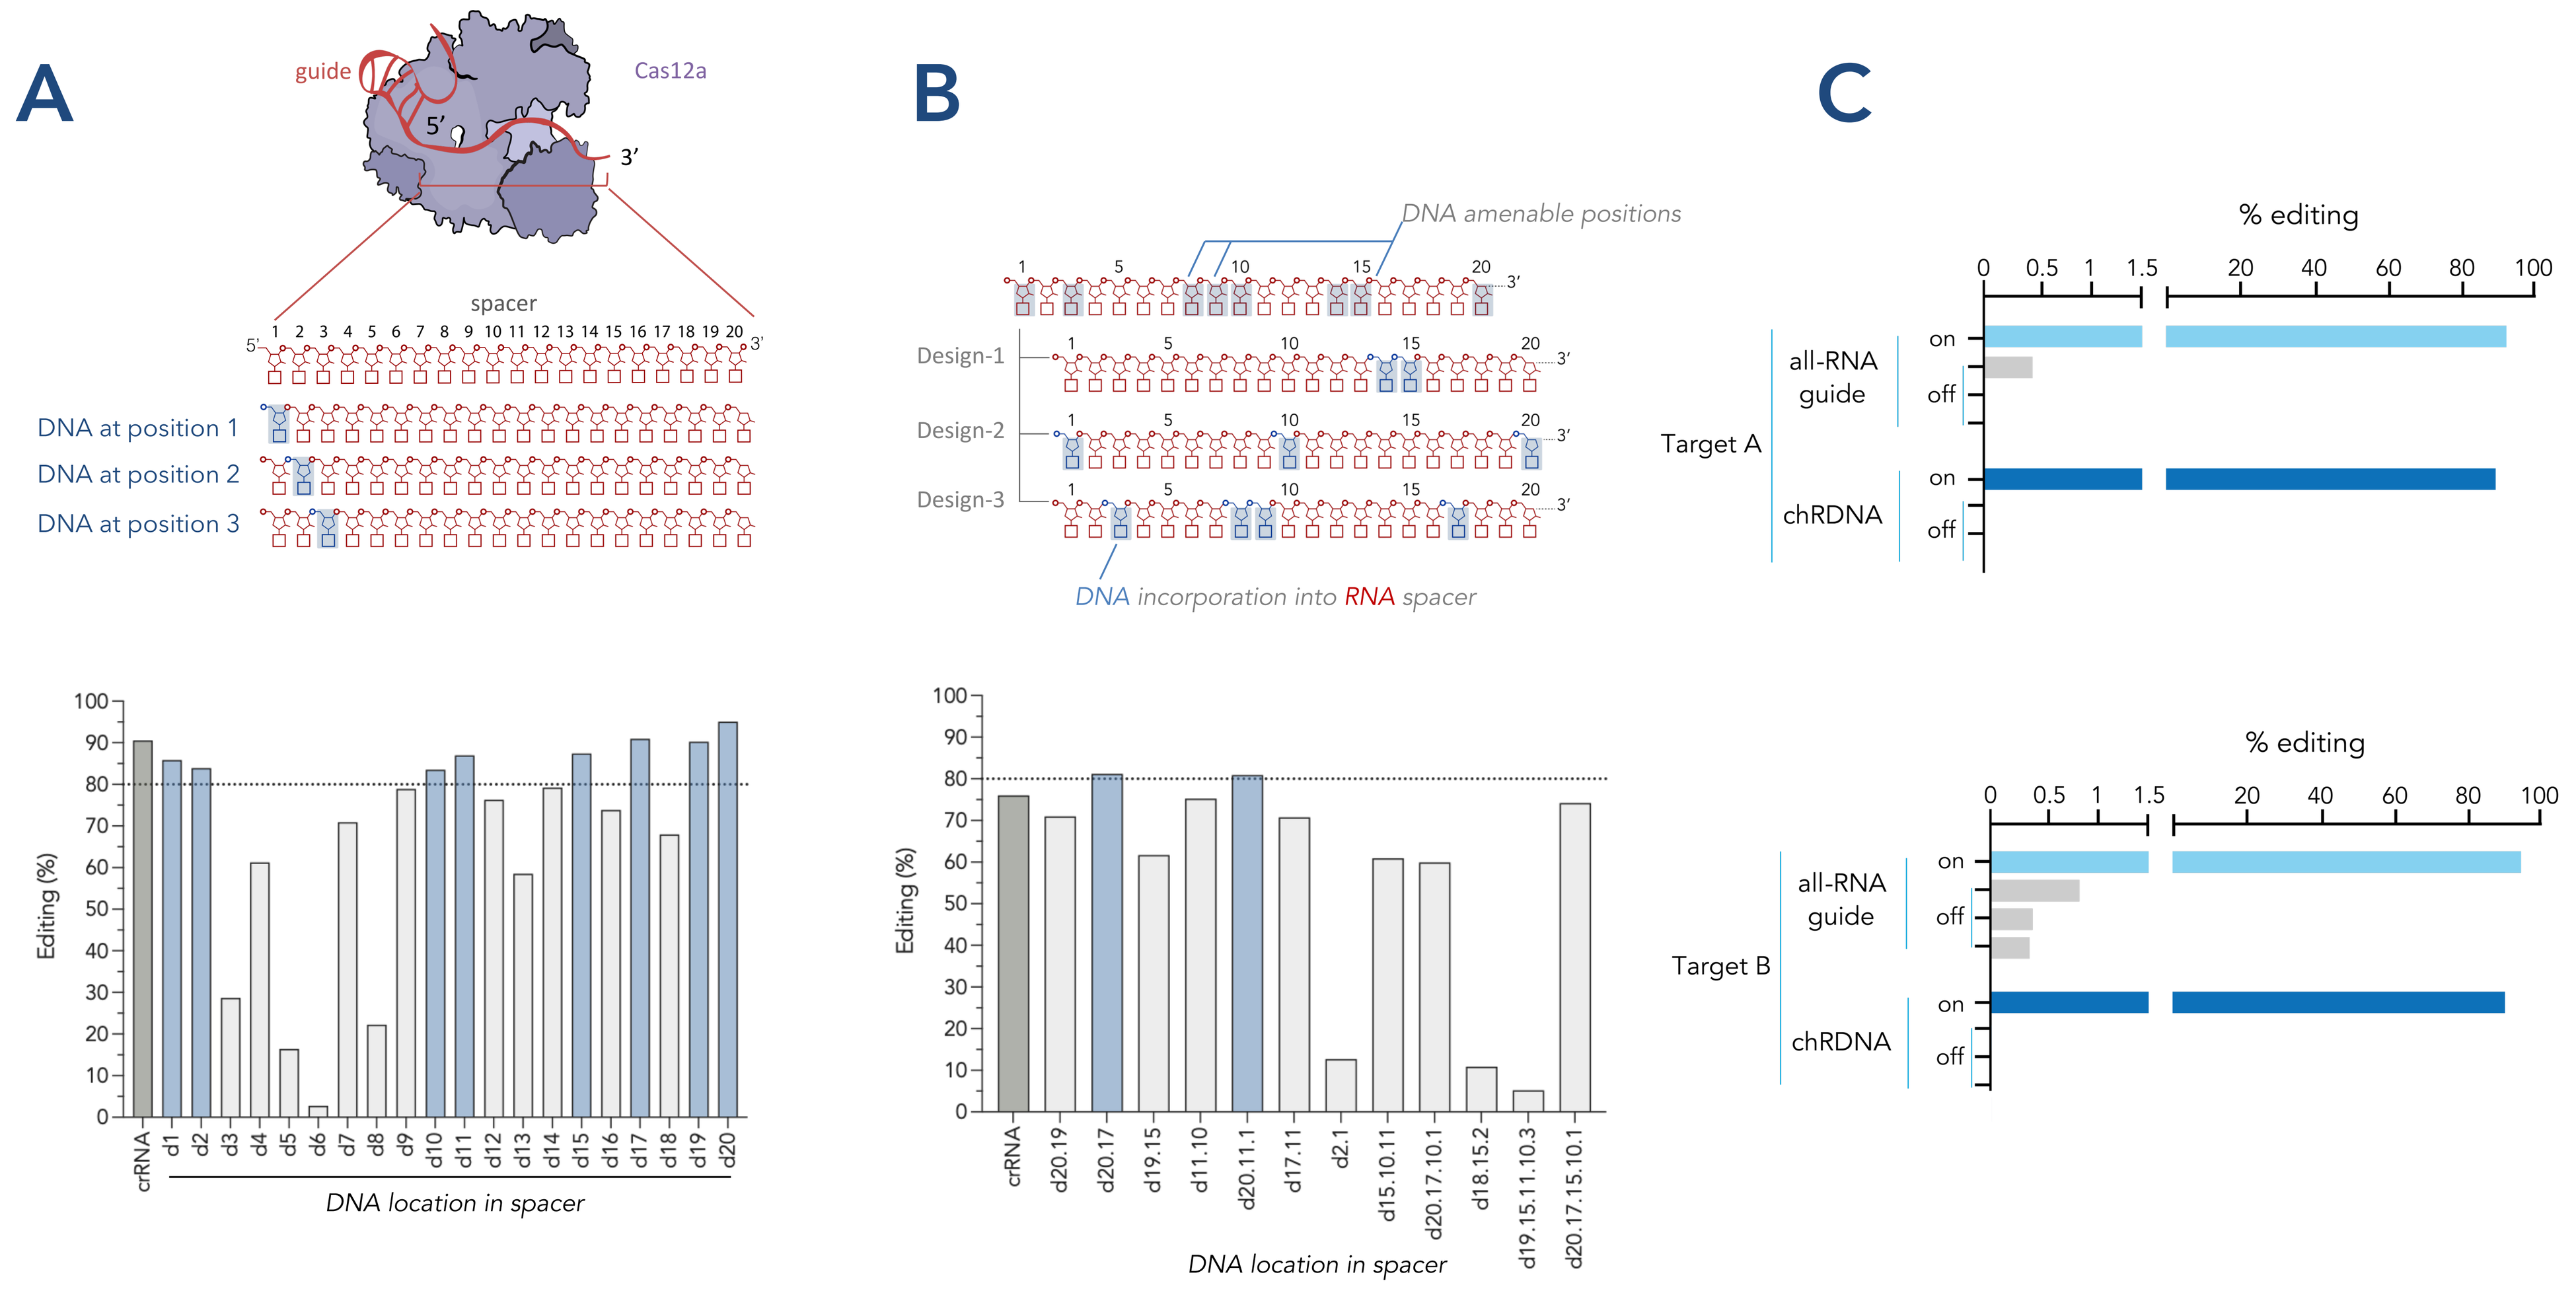
A
guide
Cas12a
spacer
DNA at position 1
DNA at position 2
DNA at position 3
Editing (%)
DNA location in spacer
B
DNA amenable positions
Design-1
Design-2
Design-3
DNA incorporation into RNA spacer
Editing (%)
DNA location in spacer
C
% editing
Target A
all-RNA guide
chRDNA
on
off
Target B

Enhancing Cas12a editing levels through optimization of NLS sequence

Knowing that the manufacturing of single- and multi-plex genome-edited cell therapies would benefit from consistent editing frequencies, we endeavored to enhance the editing capability of our Cas12a platform through optimization of the nuclear localization signal sequence (NLS) appended to the c-terminus of the nuclease. We screened a matrix of designs evaluating the type and number of NLSs as well as the linker length and composition used to append the NLS to the Cas12a. We found that multiple designs afforded improved editing levels across numerous targets compared to the standard gly-ser linker and SV40 NLS sequence (A). Selecting top targets from (A), we compared the editing in a time course assays and showed that the optimized NLS resulted in a more rapid accumulation of edits in T cells, perhaps indicating more rapid nuclear trafficking (B).

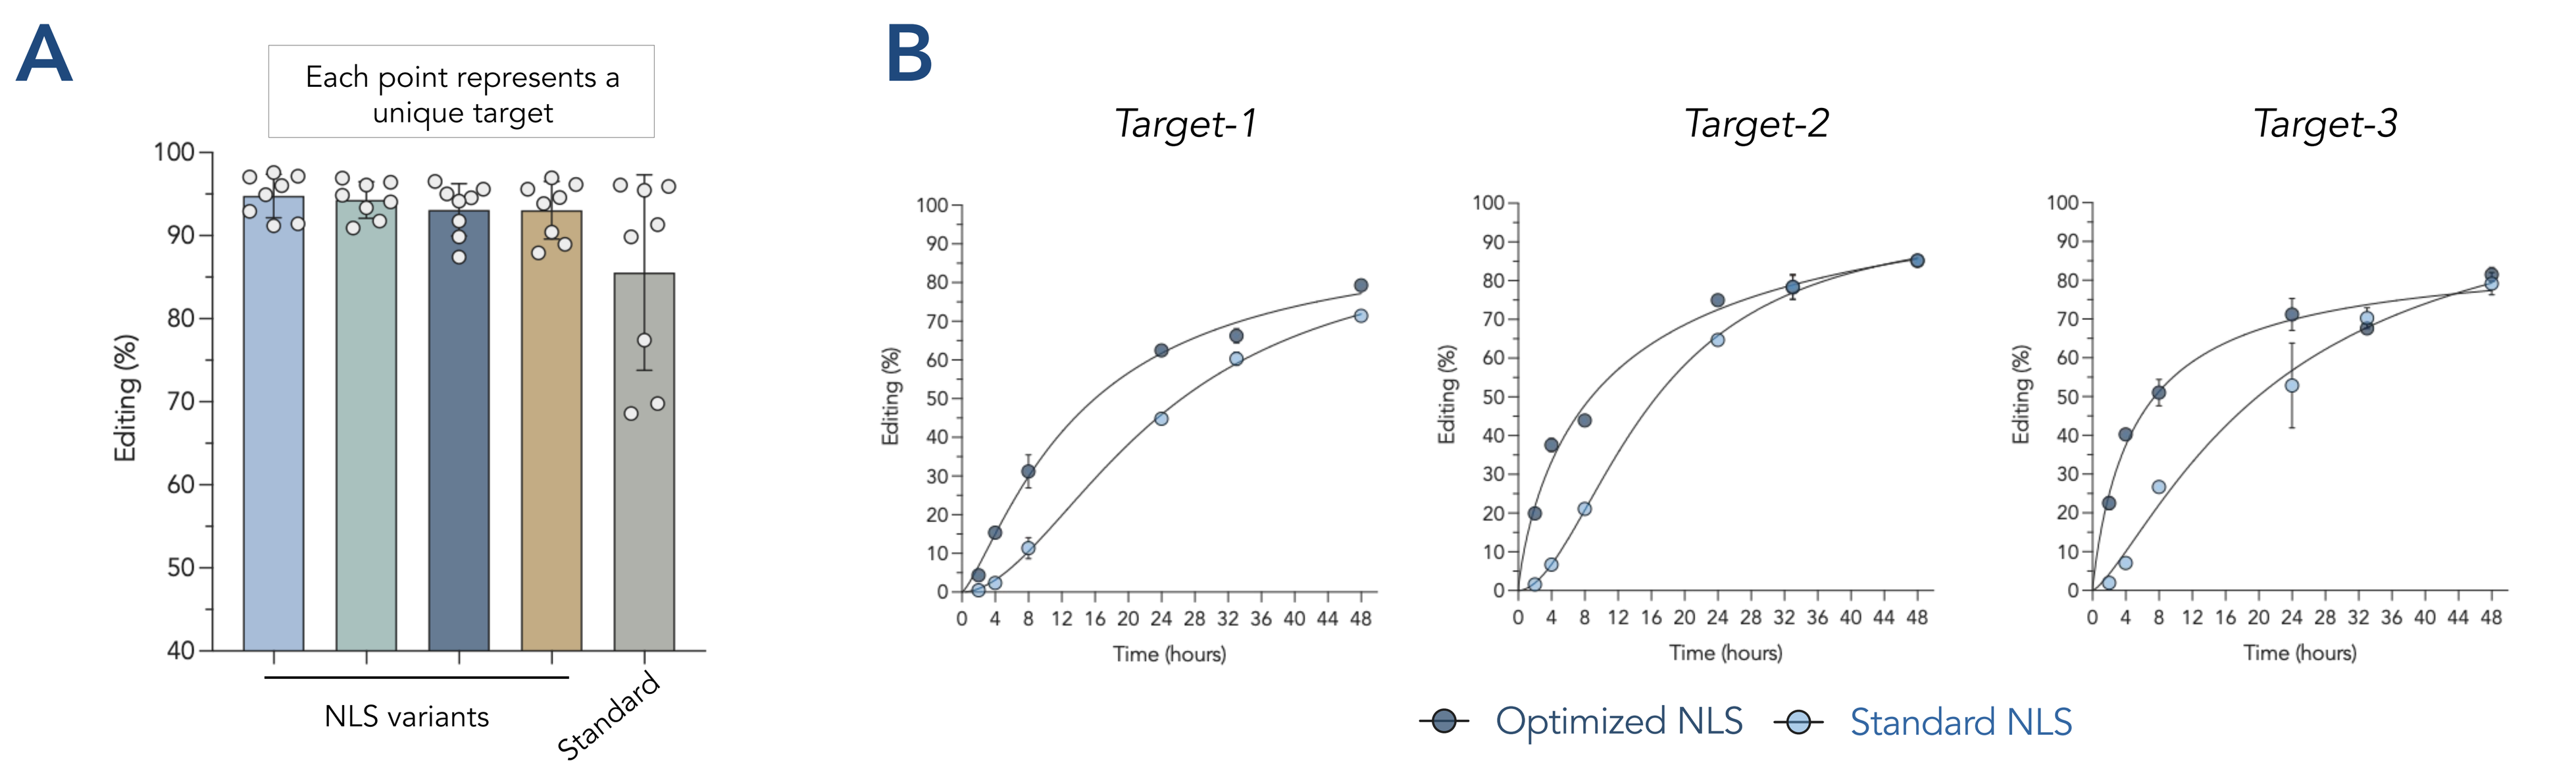
A
Each point represents a unique target
Editing (%)
NLS variants
Standard
B
Target-1
Target-2
Target-3
Editing (%)
Time (hours)
Optimized NLS
Standard NLS

Optimized Cas12a-NLS improves sequential editing rates in iPSCs

To confirm the utility of the optimized Cas12a-NLS variant we utilized it for sequential editing in induced pluripotent stem cells (iPSCs). iPSCs were edited in a step-wise fashion at four genes with either the standard or optimized NLS and at the end of the four editing rounds, cells were sequenced at all four loci (A). To determine the theoretical fraction of cells in the polyclonal population that contain all four edits, we multiplied the four individual gene editing frequencies together. For the standard NLS, we determined that 1 out of every 9 cells would likely contain all four desired edits but that the optimized NLS yielded a population of cells where half of the cells would contain all edits, a 5-fold improvement in efficiency (B).

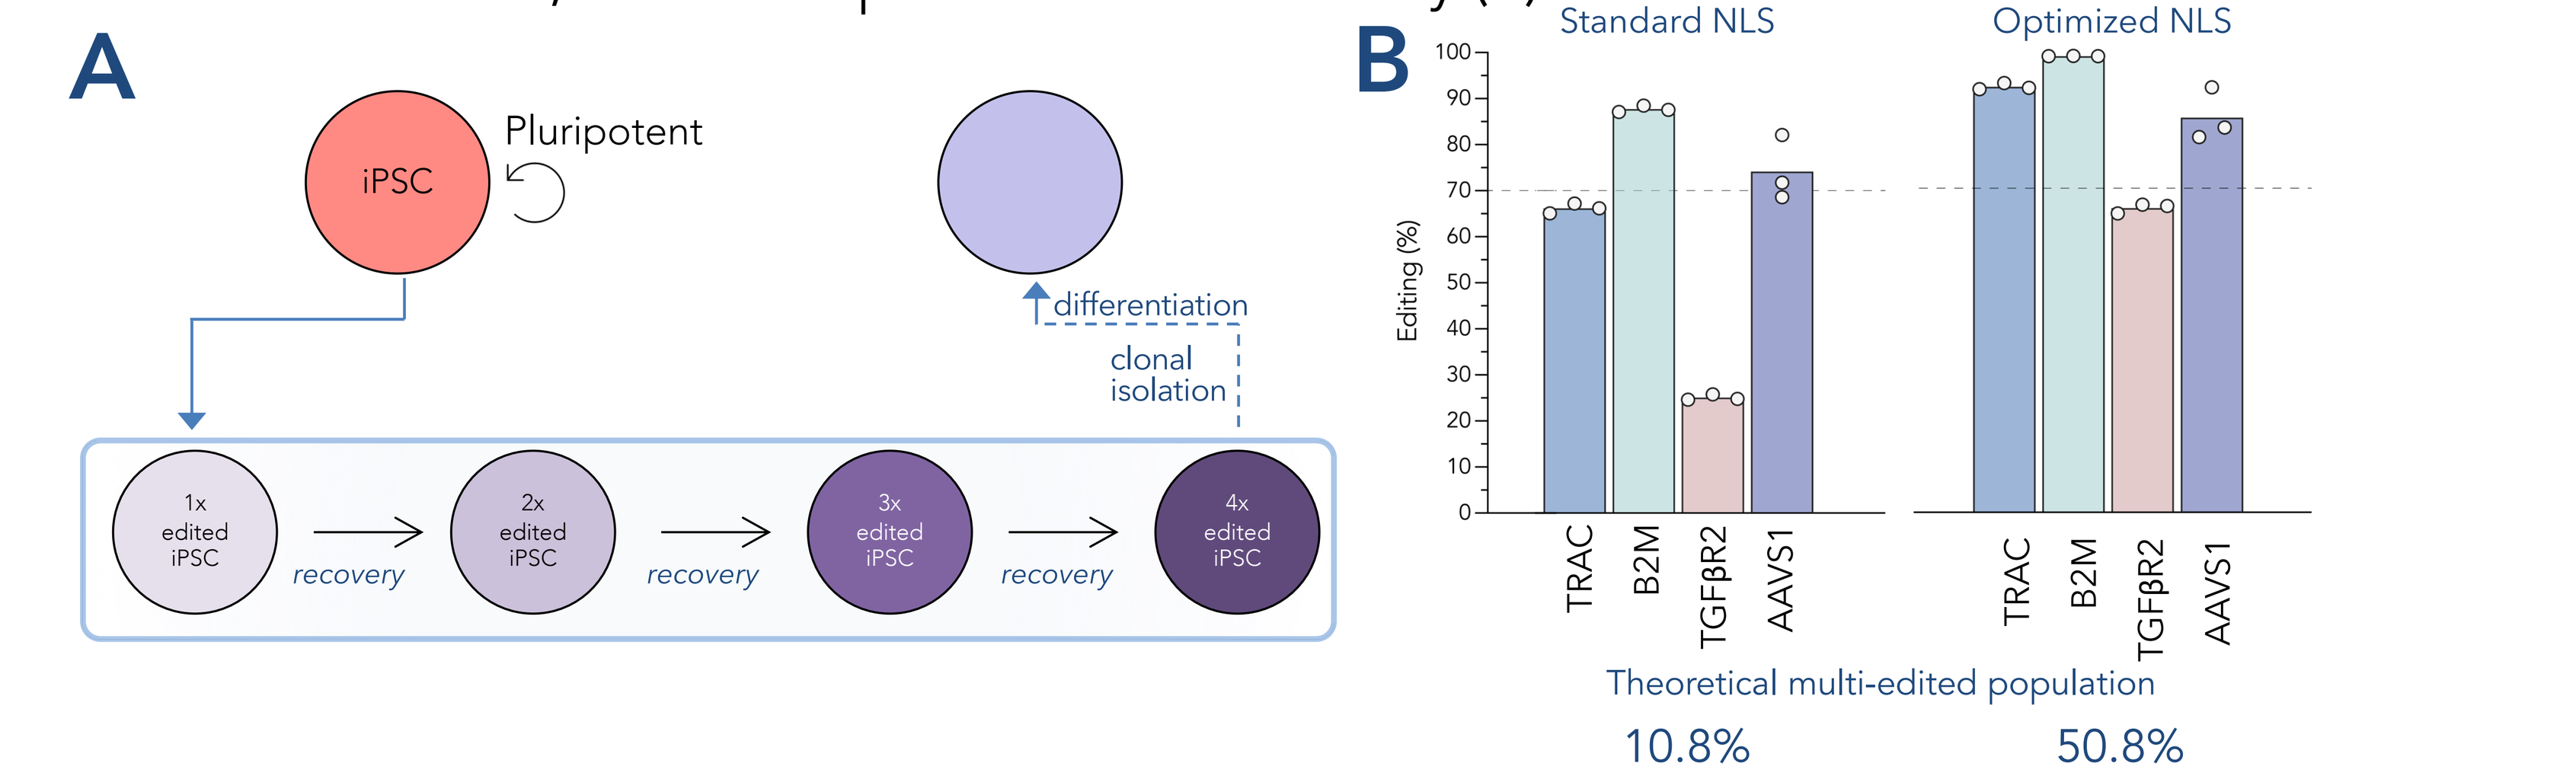
A
iPSC
Pluripotent
differentiation
clonal isolation
1x edited iPSC
2x edited iPSC
3x edited iPSC
4x edited iPSC
recovery
B
Standard NLS
Optimized NLS
Editing (%)
TRAC
B2M
TGFβR2
AAVS1
Theoretical multi-edited population
10.8%
50.8%

Cas12a achieves robust editing across multiple immune cell types

To better understand the broad utility of the optimized Cas12a-NLS beyond T and iPS cells, we utilized Cas12a editing in human iNK cells, primary B cells, and macrophages achieving ≥80% on-target editing across multiple gene targets.

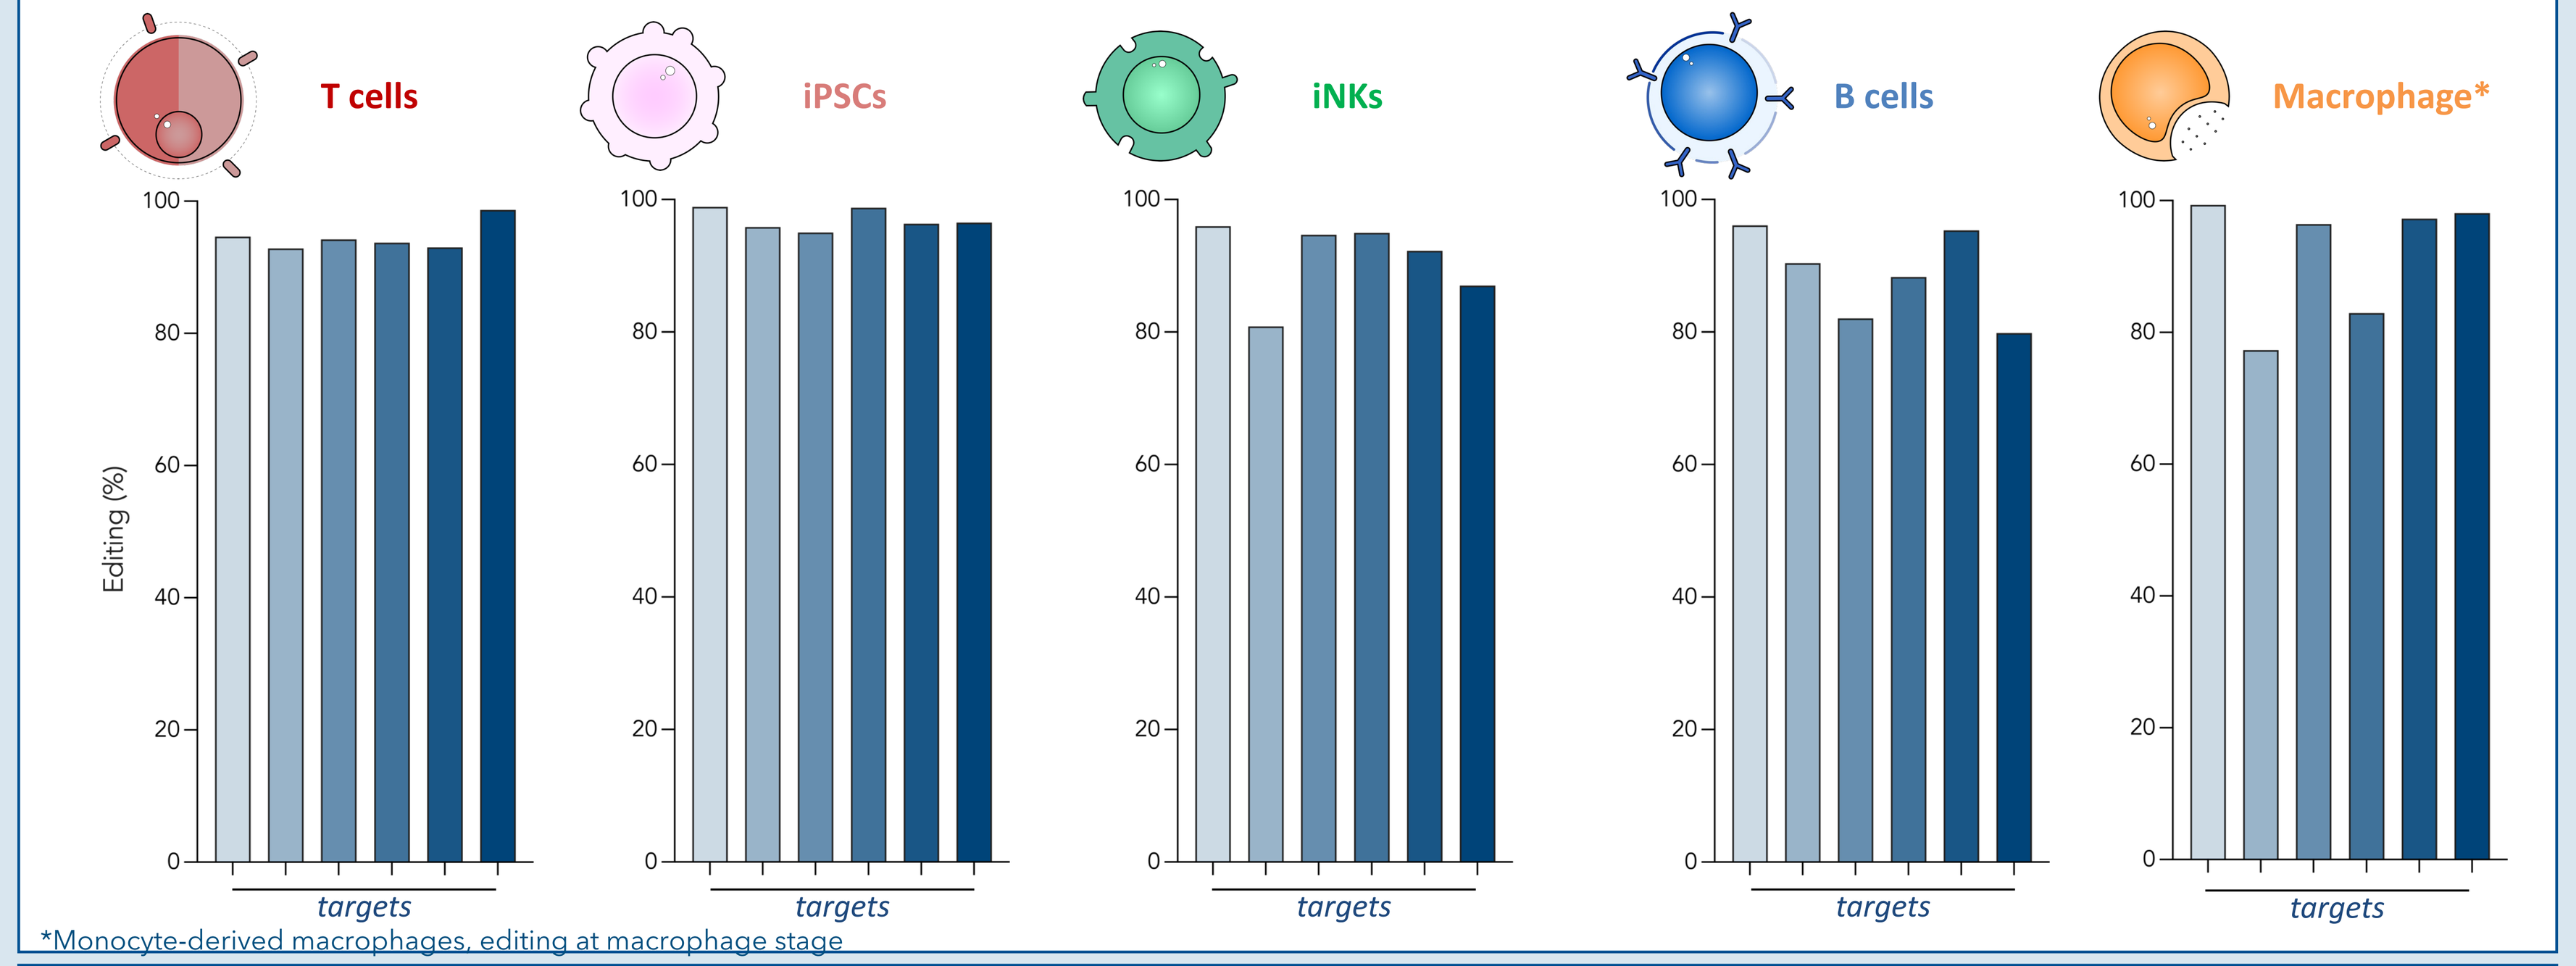
T cells
iPSCs
iNKs
B cells
Macrophage*
Editing (%)
targets

*Monocyte-derived macrophages, editing at macrophage stage

Summary

- Cas9 and Cas12a chRDNAs demonstrate improved specificity compared to all-RNA guides, while maintaining robust on-target editing rates
- Editing efficiency of Cas12a chRDNA is further enhanced through utilization of an optimized NLS, resulting in faster target cleavage and improved yields of multi-edited cell populations
- Cas12a chRDNAs demonstrate broad and robust editing across multiple immune cell types
- Caribou utilizes both Cas9 and Cas12a chRDNAs across three clinical-stage CAR T cell programs CB-010 (NCT04637763), CB-011 (NCT05722418), and CB-012 (NCT06128044) for the treatment of patients with hematologic malignancies

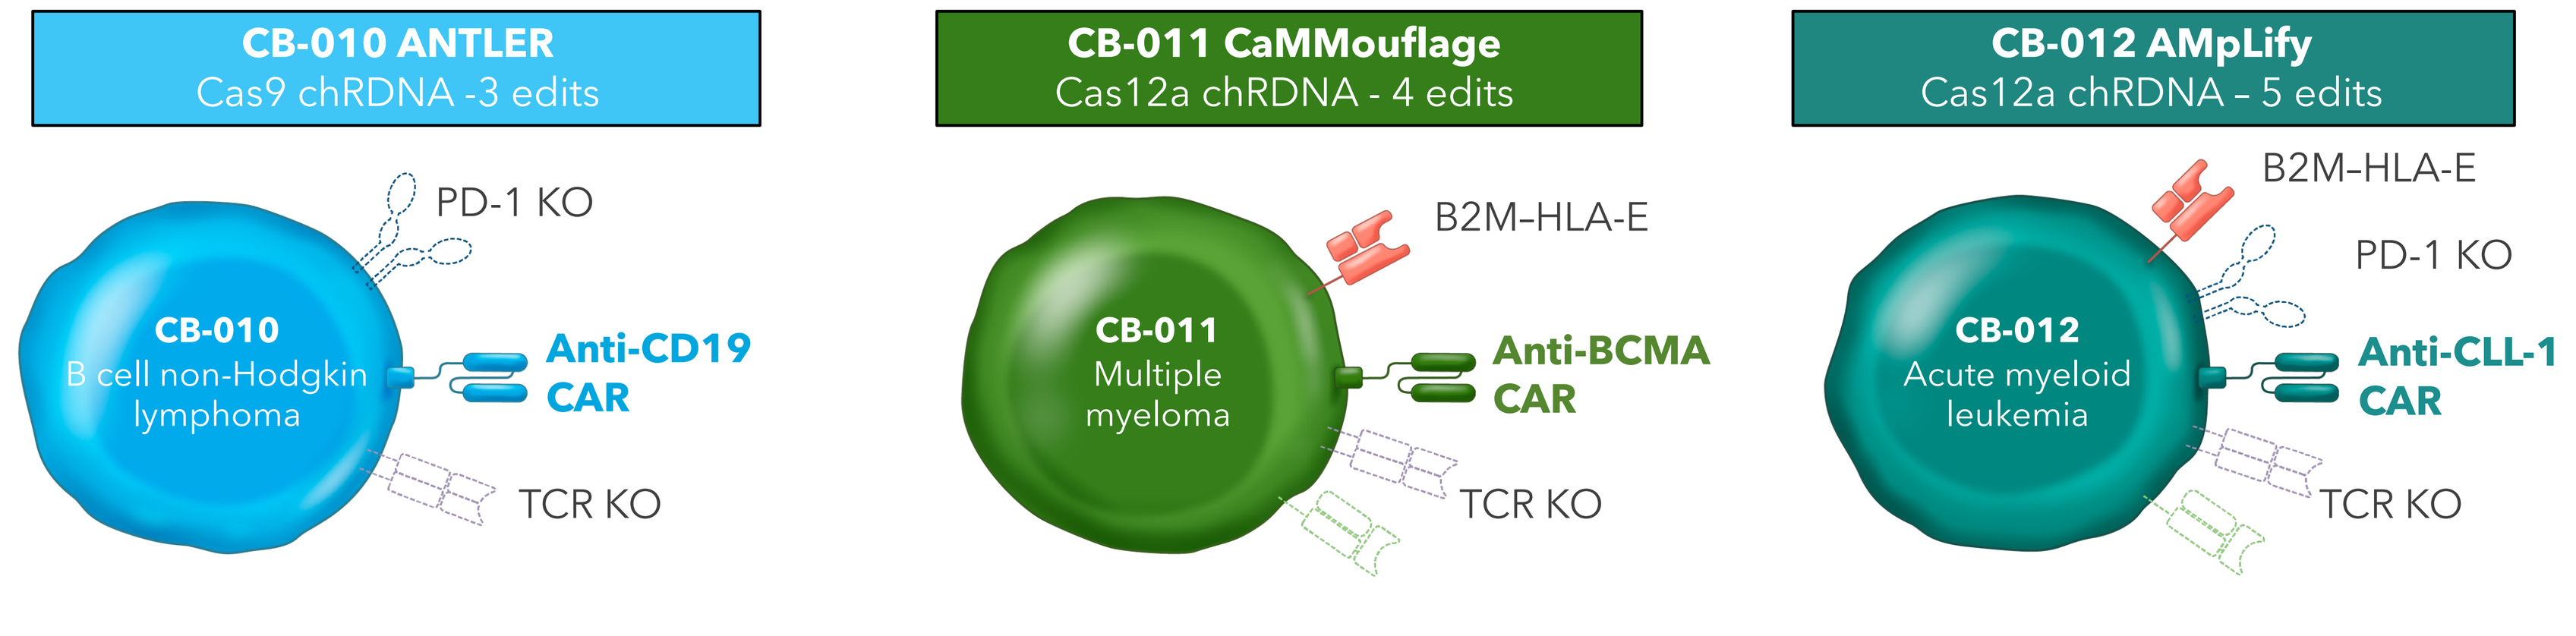
CB-010 ANTLER
Cas9 chRDNA -3 edits
CB-011 CaMMouflage
Cas12a chRDNA - 4 edits
CB-012 AMpLify
Cas12a chRDNA – 5 edits
CB-010
B cell non-Hodgkin lymphoma
PD-1 KO
Anti-CD19 CAR
TCR KO
CB-011
Multiple myeloma
B2M-HLA-E
Anti-BCMA CAR
TCR KO
CB-012
Acute myeloid leukemia
B2M-HLA-E
PD-1 KO
Anti-CLL-1 CAR
TCR KO

TANDEM MEETINGS: Transplantation & Cellular Therapy Meetings of ASTCT™ and CIBMTR®
February 21-24, 2023 | San Antonio, Texas

CORRESPONDING AUTHOR: PAUL DONOHOUE (donohoue@cariboubio.com).

© 2024 Caribou Biosciences, Inc. "Caribou Biosciences" and Caribou's logo are registered trademarks of Caribou Biosciences, Inc.